P326

Safety of Ocrelizumab in Multiple Sclerosis: Updated Analysis in Patients with Relapsing and Primary Progressive Multiple Sclerosis

SL Hauser,[1] L Kappos,[2] X Montalban,[3] C Chognot,[4] N Jessop,[4] K Kadner,[4] A Pradhan,[5] K Prajapati,[6] JS Wolinsky[7]

[1]UCSF Weill Institute for Neurosciences, University of California San Francisco, San Francisco, CA, USA; [2]Research Center for Clinical Neuroimmunology and Neuroscience, University Hospital Basel, University of Basel, Basel, Switzerland; [3]Department of Neurology-Neuroimmunology, Multiple Sclerosis Centre of Catalonia, Vall d'Hebron University Hospital, Barcelona, Spain; [4]F. Hoffmann-La Roche Ltd, Basel, Switzerland; [5]Genentech, Inc., South San Francisco, CA, USA; [6]IQVIA Solutions, Inc., Amsterdam, The Netherlands; [7]McGovern Medical School, The University of Texas Health Science Center at Houston (UTHealth), Houston, TX, USA

The aim of this study was to demonstrate the continuity in safety of ocrelizumab (OCR) by reporting longer-term safety evaluations from OCR clinical trials and open-label extension periods over a 9-year follow-up (up to November 2021)

RESULTS

Nov 2021

As of November 2021, 5,848 patients in the OCR all-exposure population (patients with multiple sclerosis [MS] across multiple clinical trials) had received OCR (amounting to 25,153 patient years of exposure); approximately 50% of patients (2,836) received at least 8 doses

Mar 2022

As of March 2022, over 250,000 patients with MS had started OCR globally in the post-marketing setting

AEs With and Without COVID-19 Cases. Over 9 Years, the Safety Profile of OCR Remained Consistent

Adverse event Rate per 100 PY (95% CI)	CTP[a] OPERA IFN β-1a	CTP[a] OPERA OCR	CTP[a] ORATORIO Placebo	CTP[a] ORATORIO OCR	CTP/OLE[b] OPERA OCR	CTP/OLE[b] OPERA (Ex-COVID-19) OCR	CTP/OLE[b] ORATORIO OCR	CTP/OLE[b] ORATORIO (Ex-COVID-19) OCR	OCR all-exposure population[c]	Excl. COVID-19 cases
Total no. of patients	826	825	239	486	1,448	1,448	644	644	5,848	5,848
Total PY	1,399	1,448	729	1,606	9,890.8	9,890.8	4,336.4	4,336.4	25,153.0	25,153.0
Any AEs	**296** (287–305)	**290** (281–299)	**259** (247–271)	**252** (244–260)	**199.64** (196.90–202.40)	**196.91** (194.15–199.70)	**225.30** (220.86-229.81)	**224.00** (219.51–228.44)	**233** (231–235)	**230** (228–232)
AEs leading to discontinuation	**3.93** (2.96–5.12)	**2.35** (1.63–3.28)	**1.10** (0.47–2.16)	**1.25** (0.76–1.92)	**1.23** (1.02–1.47)	**1.18** (0.98–1.42)	**1.01** (0.74–1.36)	**1.01** (0.74–1.36)	**0.97** (0.85–1.10)	**0.93** (0.81–1.06)
Serious AEs	**6.3** (5.1–7.8)	**5.4** (4.3–6.7)	**12.1** (9.7–14.9)	**10.2** (8.7–11.8)	**6.90** (6.40–7.50)	**6.11** (5.63–6.61)	**13.08** (12.02-14.20)	**12.52** (11.49–13.62)	**7.61** (7.27–7.96)	**6.90** (6.60–7.20)
Infections and infestations	**67.8** (63.5–72.2)	**84.5** (79.9–89.4)	**72.5** (66.5–79.0)	**70.8** (66.8–75.0)	**69.7** (68.1–71.4)	**67.1** (65.5–68.7)	**72.7** (70.1–75.4)	**70.5** (68.1–73.1)	**69.9** (68.9–70.9)	**67.4** (66.4–68.4)
Serious infections[d]	**1.79** (1.16–2.64)	**0.83** (0.43–1.45)	**3.02** (1.89–4.57)	**2.74** (1.99–3.68)	**2.45** (2.15–2.78)	**1.62** (1.38–1.89)	**4.89** (4.25–5.59)	**4.34** (3.74–5.00)	**2.74** (2.54–2.96)	**2.04** (1.87–2.22)
IRRs	**7.9** (6.5–9.5)	**34.9** (31.9–38.1)	**20.3** (17.2–23.8)	**31.0** (28.3–33.9)	**11.91** (11.24–12.61)	**11.91** (11.24–12.61)	**17.6** (16.4–18.9)	**17.6** (16.4-18.9)	**23.0** (22.4–23.6)	**23.0** (22.4–23.6)
Malignancies[e,f]	**0.14** (0.02–0.52)	**0.28** (0.08–0.71)	**0.27** (0.03–0.99)	**0.93** (0.52–1.54)	**0.43** (0.31–0.59)	**0.43** (0.31–0.59)	**1.01** (0.74–1.36)	**1.01** (0.74–1.36)	**0.47** (0.39–0.57)	**0.47** (0.39–0.57)
Deaths	**0.14** (0.02–0.52)	**0.07** (0.00–0.38)	**0.41** (0.08–1.20)	**0.25** (0.07–0.64)	**0.23** (0.15–0.35)	**0.07** (0.03–0.15)	**0.51** (0.32–0.77)	**0.44** (0.26–0.68)	**0.30** (0.24–0.38)	**0.16** (0.12–0.22)

AE rates in the OCR all-exposure population remain generally consistent with the CTP in RMS/PPMS populations
Excluding COVID-19, the rate of SIs remained low and within the range reported in real-world registries[1]
The COVID-19 pandemic resulted in an increase in the number of cases of infections related to SARS-CoV-2 during the reporting interval

In the Ex-COVID-19 analysis, patients continued to contribute to the incidence of all other AEs. AEs were classified according to MedDRA versions 18.0, 18.1, 22.1 and 24.1. Multiple occurrences of the same AE in one patient are counted multiple times, except for malignancies.
[a]Data as of April–July 2015; [b]Includes patients who received any dose of OCR during the CTP and associated OLE periods of the Phase III studies, including patients originally randomised to comparator (IFN β-1a or placebo) who switched to open-label OCR treatment (data as of November 2021); [c]Includes patients who received any dose of OCR during the CTP and associated OLE periods of the Phase II and Phase III studies plus VELOCE, CHORDS, CASTING, OBOE, ENSEMBLE, LIBERTO and CONSONANCE, including patients originally randomised to comparator (IFN β-1a or placebo) who switched to open-label OCR treatment (data as of November 2021); [d]Serious infections are defined using AEs falling into the MedDRA SOC 'Infections and Infestations', and using 'Is the event nonserious or serious?' from the AE case report form; [e]Malignancies are identified using AEs falling into the standard MedDRA query 'Malignant tumours (narrow)'; [f]For malignancies, incidence rates are reported and exposure in PY was calculated from first treatment to onset of first malignancy.
AE, adverse event; CI, confidence interval; CTP, controlled treatment period; Ex, excluding; IFN, interferon; IRR, infusion-related reaction; MedDRA, Medical Dictionary for Regulatory Activities; OCR, ocrelizumab; OLE, open-label extension;
PPMS, primary progressive multiple sclerosis; PY, patient years; RMS, relapsing multiple sclerosis; SARS-CoV-2, severe acute respiratory syndrome coronavirus 2; SI, serious infection; SOC, System Organ Class.

Excluding COVID-19 Infections, No New or Particular Patterns of SIs Were Identified by Year in Either the (A) RMS or (B) PPMS OCR-Treated Populations

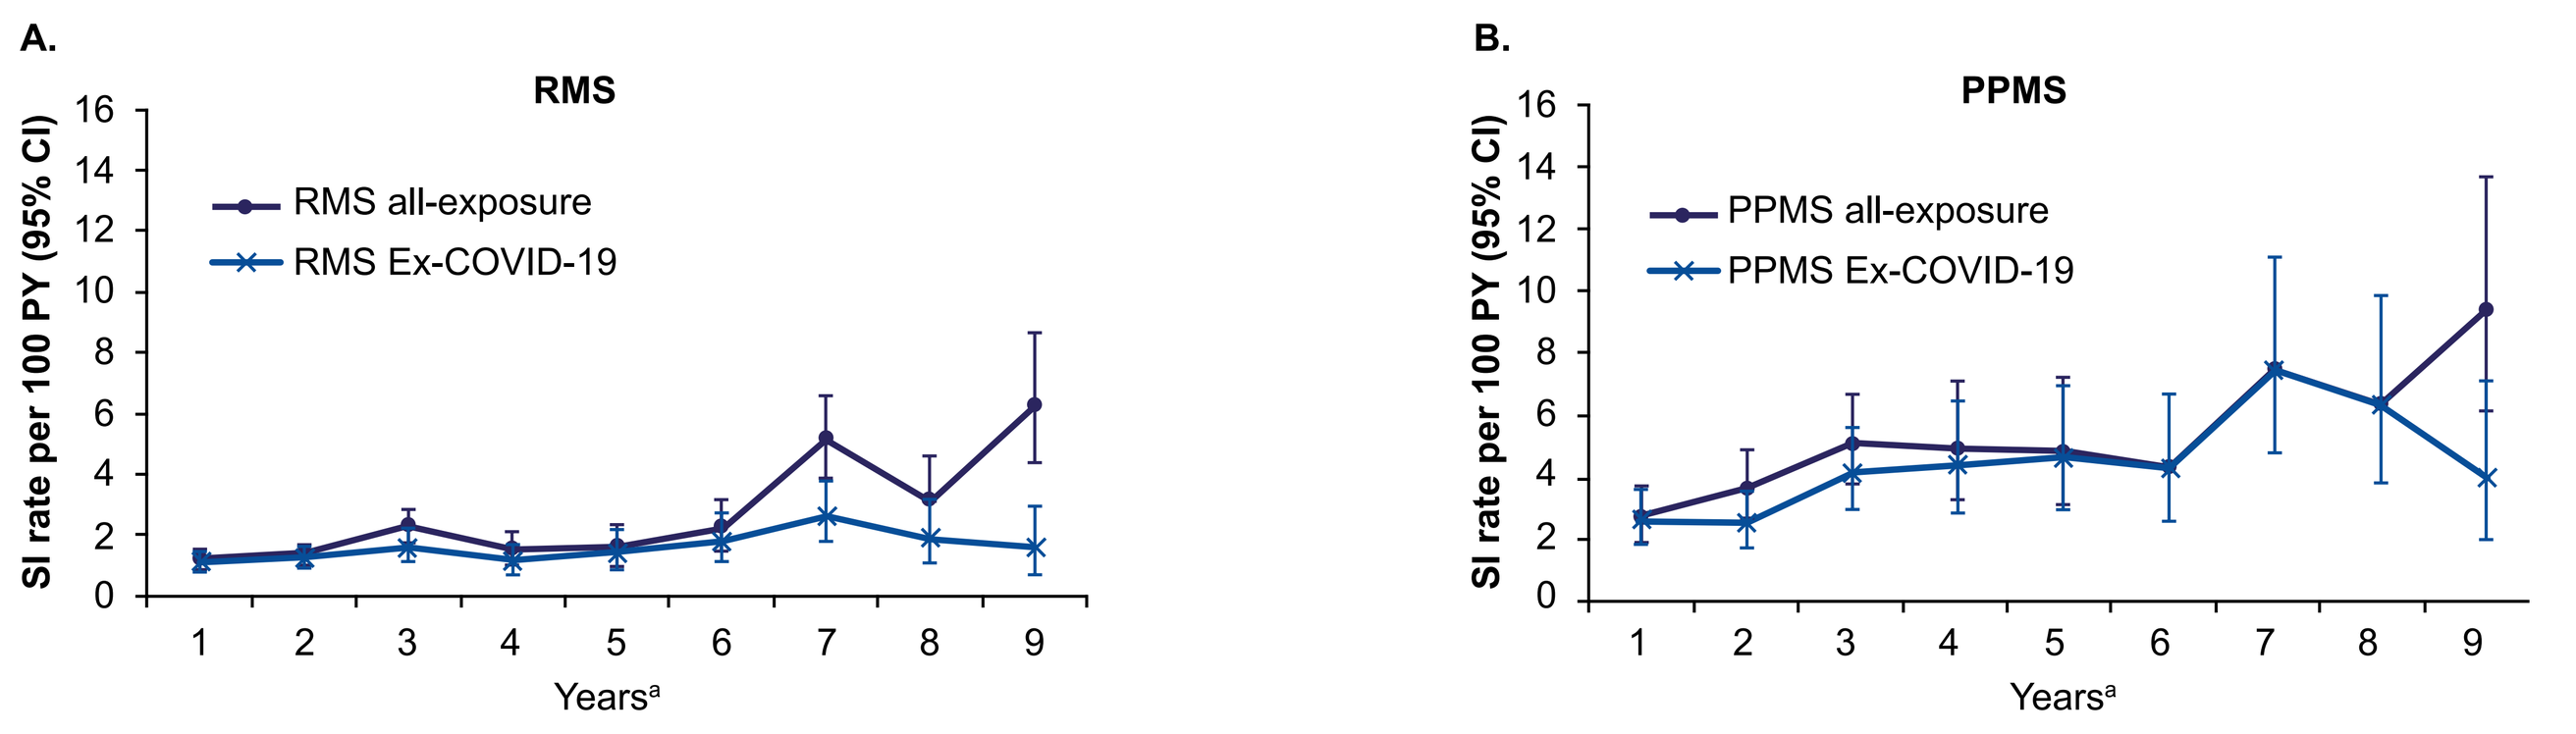
A.
RMS
SI rate per 100 PY (95% CI)
RMS all-exposure
RMS Ex-COVID-19
Years[a]
B.
PPMS
SI rate per 100 PY (95% CI)
PPMS all-exposure
PPMS Ex-COVID-19
Years[a]

A. RMS

Year	1	2	3	4	5	6	7	8	9
Exposure (PY)	4,320 4,320	4,085 4,085	2,968 2,968	2,214 2,214	1,414 1,414	1,230 1,230	1,138 1,138	779 779	576 576
Event rate (per 100 PY)	1.16 1.13	1.32 1.25	2.26 1.58	1.54 1.13	1.56 1.41	2.20 1.79	5.10 2.64	3.08 1.93	6.25 1.56
Number of SIs	50 49	54 51	67 47	34 25	22 20	27 22	58 30	24 15	36 9
N	4,376 4,376	4,257 4,257	3,428 3,428	2,498 2,498	1,593 1,593	1,291 1,291	1,198 1,198	965 965	642 642

B. PPMS

Year	1	2	3	4	5	6	7	8	9
Exposure (PY)	1,345 1,345	1,235 1,235	971 971	571 571	497 497	443 443	322 322	300 300	278 278
Event rate (per 100 PY)	2.68 2.60	3.65 2.51	5.05 4.12	4.90 4.38	4.83 4.63	4.29 4.29	7.45 7.45	6.33 6.33	9.35 3.95
Number of SIs	36 35	45 31	49 40	28 25	24 23	19 19	24 24	19 19	26 11
N	1,472 1,472	1,272 1,272	1,187 1,187	654 654	532 532	472 472	343 343	310 310	293 293

Overall, the most frequently reported SIs were consistent with those typically reported for each year
In PPMS, the rate of SIs remained higher than RMS; over time, this could be due to the underlying disease condition (e.g. increasing disability, age, comorbidities)
The majority of SIs were typical in character, resolved and were not treatment limiting

In the Ex-COVID-19 analysis, patients continued to contribute to the incidence of all other AEs.[a]The exposure in PY during Year 9 is limited for meaningful interpretation.
CI, confidence interval; Ex, excluding; PPMS, primary progressive multiple sclerosis; PY, patient years; RMS, relapsing multiple sclerosis; SI, serious infection.

The Majority of Infection AEs Were of Grade 3 Intensity, Had Resolved and Were Not Treatment Limiting Despite the COVID-19 Pandemic

OCR all-exposure population

AEs and outcomes
Total exposure 25,153 PY
Total AEs n=58,534

Total infections
n=17,579

Total serious infections: n=690
Serious infection rate per 100 PY (95% CI): 2.74 (2.54–2.96)

Outcomes:
84.8% (585/690) **resolved**
6.2% (43 events) **resolved with sequelae**
0.9% (6 events) resolving
1.9% (13 events) unresolved
6.2% (43 events) fatal[a]

Intensity grade (CTCAE)[b]
1: <0.1%
2: 1.2%
3: 5.8%
4: 1.0%
5: 0.7%

Action taken with OCR:
Withdrawn 4.5% (31/690 SIs)

Excluding COVID-19 cases

AEs and outcomes
Total exposure 25,153 PY
Total AEs n=57,883

Total infections
n=16,946

Total serious infections: n=513
Serious infection rate per 100 PY (95% CI): 2.04 (1.87–2.22)

Outcomes:
91.4% (469/513) **resolved**
4.7% (24 events) **resolved with sequelae**
0.4% (2 events) resolving
1.9% (10 events) unresolved
1.6% (8 events) fatal[a]

Intensity grade (CTCAE)[b]
1: <0.1%
2: 1.0%
3: 4.4%
4: 0.7%
5: 0.1%

Action taken with OCR:
Withdrawn 4.1% (21/513 SIs)

In the Ex-COVID-19 analysis, patients continued to contribute to the incidence of all other AEs. [a]Excluding COVID-19 cases, fatal SIs included pneumonia (n=2), sepsis (n=2), encephalitis (n=1), urosepsis (n=1), enterococcal infection (n=1) and pneumonia aspiration (n=1);
[b]Grade 1 (mild): Asymptomatic or mild symptoms/clinical or diagnostic observation only/intervention not indicated; Grade 2 (moderate): Minimal, local or noninvasive intervention indicated/limiting age-appropriate instrumental ADL; Grade 3 (severe): Severe or medically insignificant but not immediately life-threatening/hospitalisation or prolongation of hospitalisation indicated/disabling/limiting self-care ADL; Grade 4 (life-threatening): Life-threatening consequences/urgent intervention required; Grade 5 (death): Death related to AE (not applicable for all AEs).
ADL, activities of daily living; AE, adverse event; CI, confidence interval; CTCAE, Common Terminology Criteria for Adverse Events; Ex, excluding; OCR, ocrelizumab; PY, patient years; SI, serious infection.

Over 9 Years, the Cumulative Standardised Incidence Rates of (A) All Malignancies and (B) Female Breast Cancer Remained Within the Range Reported in Registries[2,3]

A. Incidence of all malignancies (Age- and sex-standardised)

B. Incidence of female breast cancer (Age-standardised)

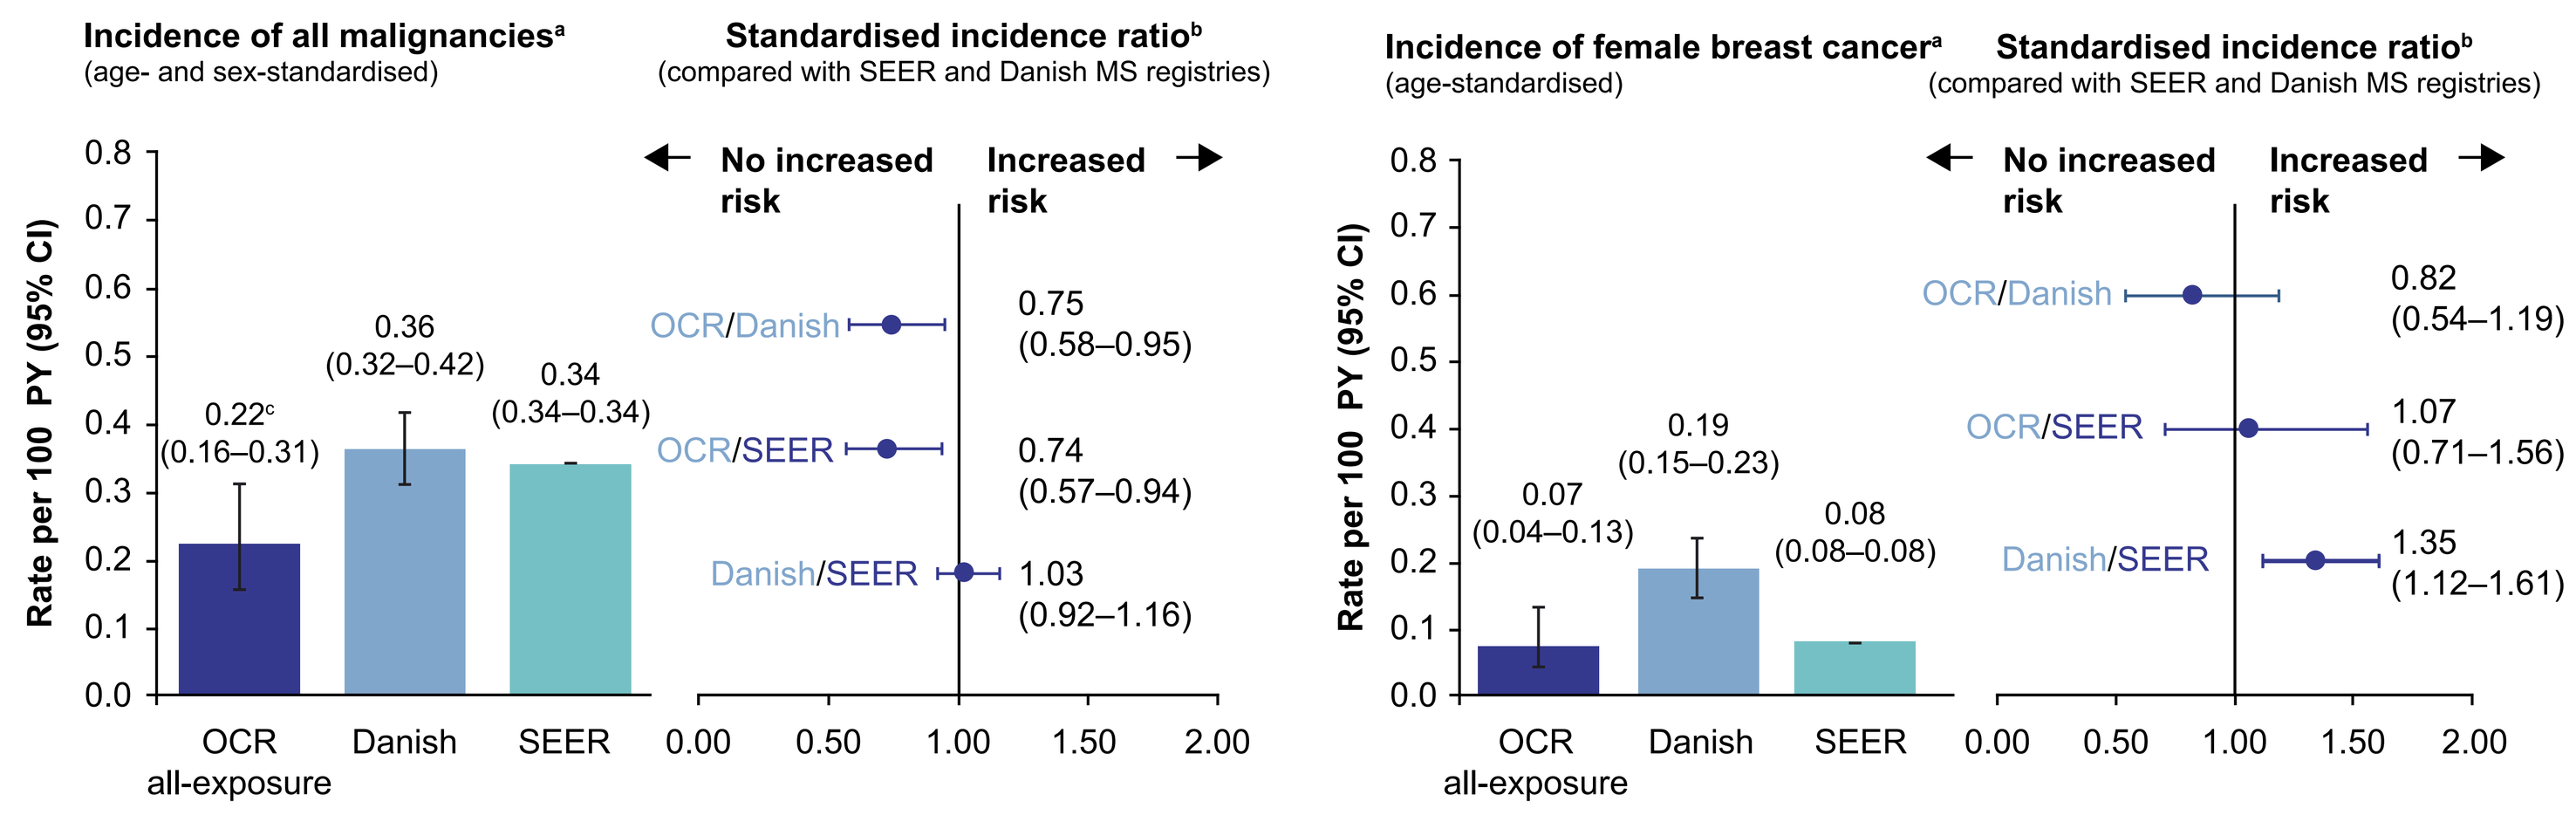
Incidence of all malignancies[a]
(age- and sex-standardised)
Standardised incidence ratio[b]
(compared with SEER and Danish MS registries)
Rate per 100 PY (95% CI)
0.22[c] (0.16–0.31)
0.36 (0.32–0.42)
0.34 (0.34–0.34)
OCR all-exposure
Danish
SEER
No increased risk
Increased risk
OCR/Danish
0.75 (0.58–0.95)
OCR/SEER
0.74 (0.57–0.94)
Danish/SEER
1.03 (0.92–1.16)
Incidence of female breast cancer[a]
(age-standardised)
Standardised incidence ratio[b]
(compared with SEER and Danish MS registries)
0.07 (0.04–0.13)
0.19 (0.15–0.23)
0.08 (0.08–0.08)
OCR/Danish
0.82 (0.54–1.19)
OCR/SEER
1.07 (0.71–1.56)
Danish/SEER
1.35 (1.12–1.61)

[a]The standardised incidence rates were reported to allow comparison with the SEER database and the Danish MS Registry, using the direct standardisation method. Standardised incidence rates were derived by applying age–sex specific rates to the 2,000 USA standard population, with restriction to the age range of the MS clinical trials (15–59 years); [b]The SIR, calculated as observed/expected number of events, was determined for all malignancies (excluding NMSC) and female breast cancer, using the SEER database and the Danish MS Registry as reference populations; [c]It excludes NMSC for comparison with SEER rates, as NMSC is not reported in SEER.
CI, confidence interval; MS, multiple sclerosis; NMSC, nonmelanoma skin cancer; OCR, ocrelizumab; PY, patient years; SEER, Surveillance, Epidemiology and End Results; SIR, standardised incidence ratio.

Withdrawals Due to AEs Over 9 Years in the OCR All-Exposure Population

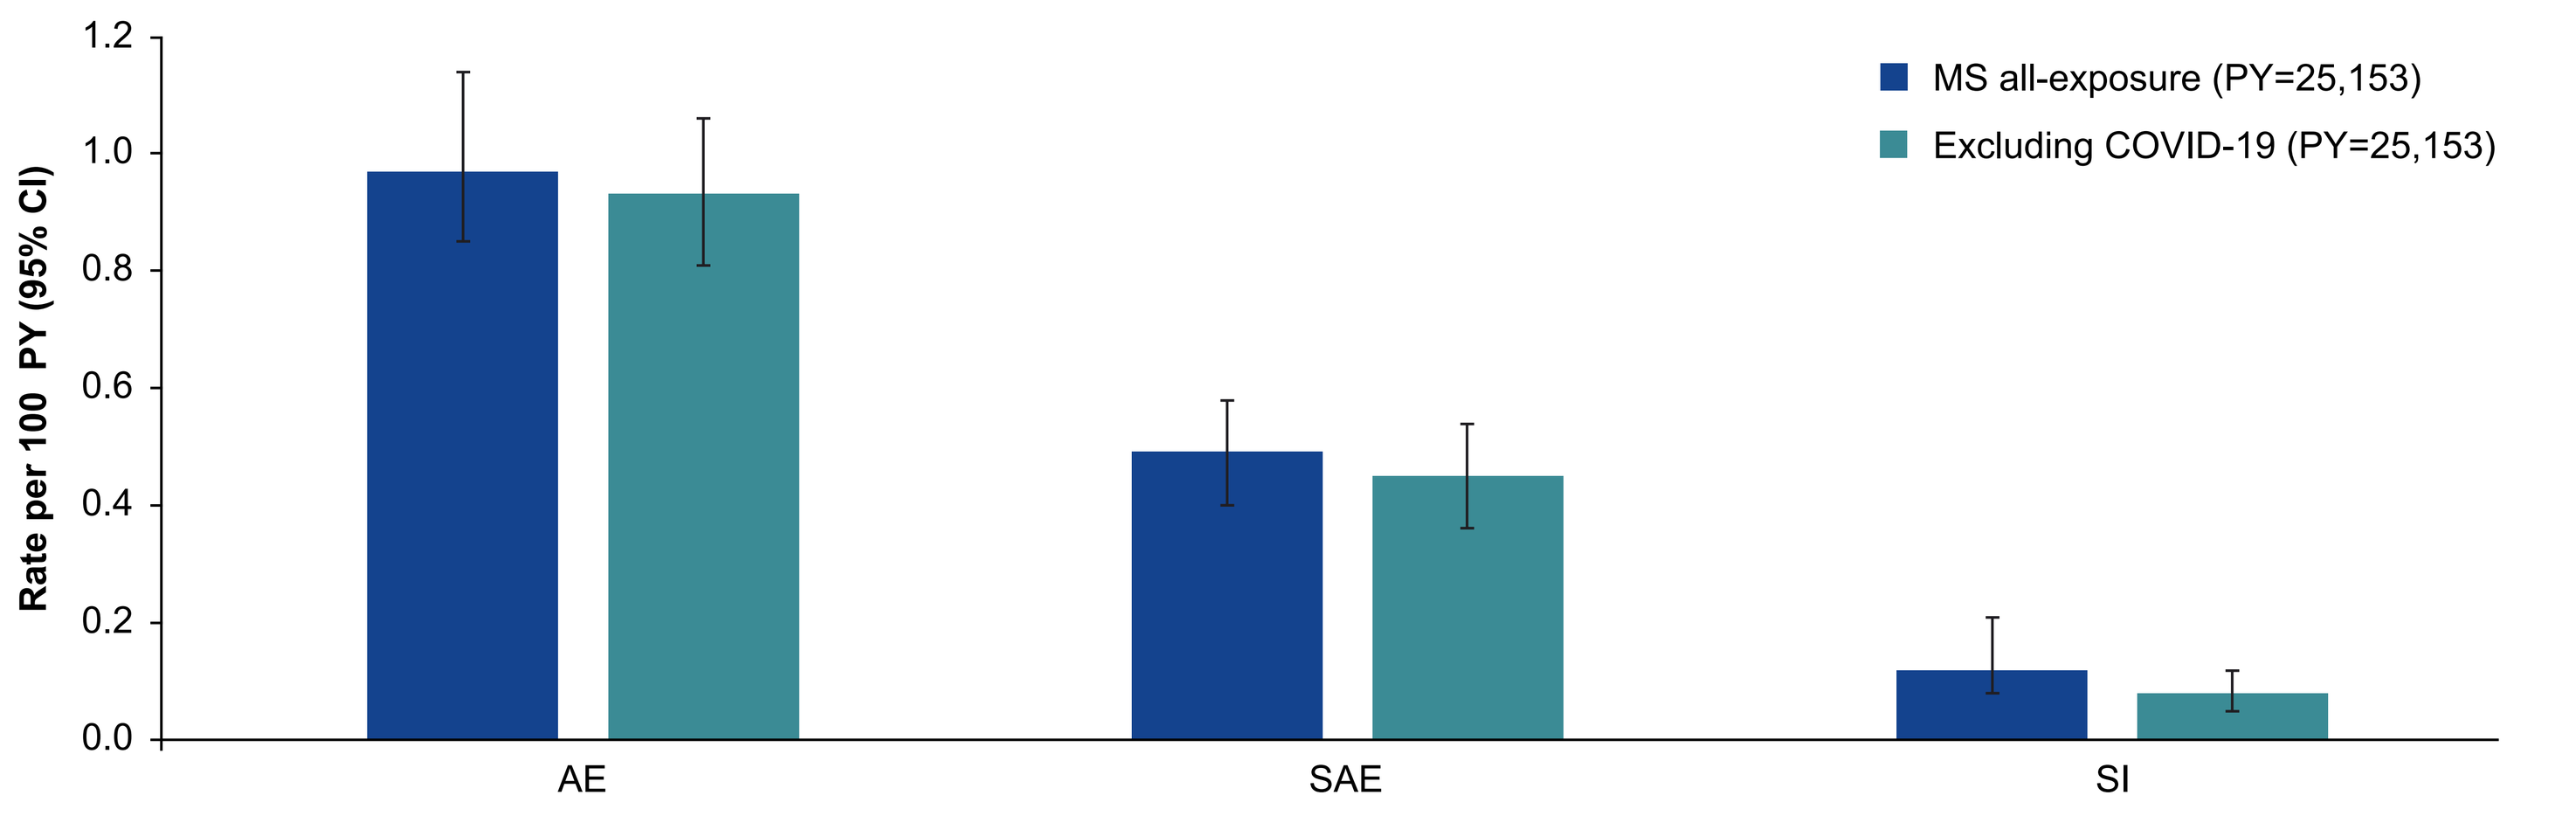
Rate per 100 PY (95% CI)
MS all-exposure (PY=25,153)
Excluding COVID-19 (PY=25,153)
AE
SAE
SI

The rate of AEs leading to patient withdrawal remained infrequent with additional exposure, despite the COVID-19 pandemic. SIs or serious COVID-19 infections are not driving the withdrawals due to AEs

In the Ex-COVID-19 analysis, patients continued to contribute to the incidence of all other AEs.
AE, adverse event; CI, confidence interval; AE, adverse event; CI, confidence interval; Ex, excluding; MS, multiple sclerosis; PY, patient years; SAE, serious adverse event; SI, serious infection.

CONCLUSIONS

- Over 9 years of treatment, AE rates in the ocrelizumab all-exposure population remain generally consistent with the controlled treatment period in RMS/PPMS populations
- Despite 2 years of the COVID-19 pandemic, the rate of AEs leading to treatment withdrawal remained infrequent and stable
- Serious infection and malignancy rates remain within the range reported for patients with MS in real-world registries
- Over a 9-year follow-up period, no new (with the exception of COVID-19 type of infections) or unexpected safety signal was seen in patients treated with ocrelizumab in ongoing clinical trials, ocrelizumab continues to exhibit a stable and favourable safety profile
- Long-term follow-up and post-marketing requirement studies will continue to monitor patient safety over time in patients with MS receiving ocrelizumab, including identified and potential risks

REFERENCES

1. Wijnands JMA, *et al. J Neurol Neurosurg Psychiatry* 2018;89:1050–1056;
2. National Institutes of Health (NIH). Surveillance, Epidemiology, and End Results Program. Available from: https://seer.cancer.gov;
3. Nørgaard M, *et al. Mult Scler Relat Disord* 2019;28:81–85.

DISCLOSURES

SL Hauser serves on the Board of Trustees for Neurona and on scientific advisory boards for Alector, Annexon and Accure; and has received travel reimbursement and writing assistance from F. Hoffmann-La Roche Ltd and Novartis for CD20-related meetings and presentations. **L Kappos'** institution (University Hospital Basel) has received the following exclusively for research support: Steering committee, advisory board and consultancy fees (Actelion, Bayer HealthCare, Biogen, Bristol Myers Squibb, Sanofi-Genzyme, Janssen, Japan Tobacco, Merck, Novartis, F. Hoffmann-La Roche Ltd, Sanofi, Santhera, Shionogi and TG Therapeutics); speaker fees (Bayer HealthCare, Biogen, Merck, Novartis, F. Hoffmann-La Roche Ltd and Sanofi); support of educational activities (Allergan, Bayer HealthCare, Biogen, CSL Behring, Desitin, Sanofi-Genzyme, Merck, Novartis, F. Hoffmann-La Roche Ltd, Pfizer, Sanofi, Shire and Teva); license fees for Neurostatus products; and grants (Bayer HealthCare, Biogen, European Union, Innosuisse, Merck, Novartis, F. Hoffmann-La Roche Ltd, Swiss MS Society and Swiss National Research Foundation). **X Montalban** has received speaking honoraria and travel expenses for participation in scientific meetings, has been a steering committee member of clinical trials or participated in advisory boards of clinical trials in the past years with AbbVie, Actelion, Alexion, Biogen, Bristol Myers Squibb/Celgene, EMD Serono, F. Hoffmann-La Roche Ltd, Immunic, Janssen, MedDay, Merck, Mylan, NervGen, Novartis, Sandoz, Sanofi-Genzyme, Teva, TG Therapeutics, ExeMed, MSIF and NMSS. **C Chognot** is an employee of F. Hoffmann-La Roche Ltd. **N Jessop** is an employee of F. Hoffmann-La Roche Ltd. **K Kadner** is an employee of F. Hoffmann-La Roche Ltd. **A Pradhan** is an employee of Genentech, Inc. **K Prajapati** has received consulting fees from F. Hoffmann-La Roche Ltd for statistical assistance, and is an employee of IQVIA Solutions, Inc. **JS Wolinsky** has received personal compensation for consulting, serving on a scientific advisory board, speaking or other activities with Avotres, Brainstorm Cell Therapeutics, Cleveland Clinic Foundation, EMD Serono, Inmagene, MedDay, Novartis/Sandoz, F. Hoffmann-La Roche Ltd/Genentech, Sanofi-Genzyme and University of Alabama; royalties are received for out-licensed monoclonal antibodies through UTHealth from Millipore Corporation.

To access this poster and supplemental material scan the QR code above or go to the following URL: https://bit.ly/3MfZ1sj

ACKNOWLEDGEMENTS

We would like to thank all patients, their families and the investigators who participated in these studies.
This study was sponsored by F. Hoffmann-La Roche Ltd, Basel, Switzerland.
Writing and editorial assistance for this presentation was provided by Articulate Science, UK, and funded by F. Hoffmann-La Roche Ltd, Basel, Switzerland.

Presented at the 38th Congress of the European Committee for Treatment and Research in Multiple Sclerosis (ECTRIMS) Amsterdam, The Netherlands and Virtual | 26–28 October 2022