Long-term safety of inavolisib (GDC-0077) in an ongoing phase 1/1b study evaluating monotherapy and in combination (combo) with palbociclib (palbo) and/or endocrine therapy in patients (pts) with *PIK3CA*-mutated, hormone receptor-positive/HER2-negative (HR+/HER2–) metastatic breast cancer (BC).

1052

Philippe L Bedard[1], Melissa K Accordino[2], Andrés Cervantes[3], Valentina Gambardella[3], Erika Hamilton[4], Antoine Italiano[5], Dejan Juric[6], Kevin Kalinsky,[7] Ian E Krop[8], Mafalda Oliveira[9], Cristina Saura[9], Peter Schmid[10], Nicholas C Turner[11], Andrea Varga[12], Noopur Shankar[13], Jennifer L Schutzman[13], Stephanie Royer-Joo[13], Miguel Valero Martin[14], Komal L Jhaveri[15]

[1]University of Toronto, Toronto, ON, Canada; [2]Columbia University Irving Medical Center, New York, NY; [3]Biomedical Research Institute INCLIVA, University of Valencia, Valencia, Spain; [4]Sarah Cannon Research Institute/Tennessee Oncology, Nashville, TN; [5]Institut Bergonié, Bordeaux, France; [6]Massachusetts General Hospital, Boston, MA; [7]Columbia University Irving Medical Center, New York, NY; [8]Dana-Farber Institute, Boston, MA (current affiliation: Yale Cancer Center, New Haven, CT); [9]Vall d'Hebron University Hospital and Vall d'Hebron Institute of Oncology (VHIO), Barcelona, Spain; [10]Centre for Experimental Cancer Medicine, Cancer Research UK Barts Centre, London, UK; [11]Royal Marsden Hospital and Institute of Cancer Research, London, UK; [12]Gustave Roussy Cancer Campus, Villejuif, France; [13]Genentech, Inc., South San Francisco, CA; [14]Roche Products Limited, Welwyn Garden City, UK; [15]Memorial Sloan Kettering Cancer Center, New York, NY

BACKGROUND AND METHODS

- Dysregulating mutations in *PIK3CA*, encoding the catalytic PI3K p110α subunit, occur in ~40% of hormone receptor-positive, HER2-negative breast cancers (HR+/HER2– BCs).[1–3] As such, the PI3K/AKT/mTOR pathway has been a frequent target of anticancer drug development.
- There are limited long-term safety data for patients treated with PI3K inhibitors, mainly due to treatment-related toxicities that resulted in sub-optimal dose intensity and/or short time-on-treatment.
- Inavolisib is a PI3Kα-specific inhibitor that also promotes degradation of mutant p110α. It has demonstrated encouraging preliminary antitumor activity in patients with *PIK3CA*-mutated HR+ BC as a monotherapy, and in combination with other anticancer agents.[4,5]
- An open-label, phase 1/1b dose-escalation study of inavolisib alone, and in combination with endocrine and targeted therapies, is ongoing (NCT03006172; GO39374);[6–10] safety data for patients on treatment ≥1 year are compared with the overall safety population (shown in blue text throughout).
- Figure 1 shows the study design and number of patients on treatment for ≥1 year per arm.

Figure 1: Study design

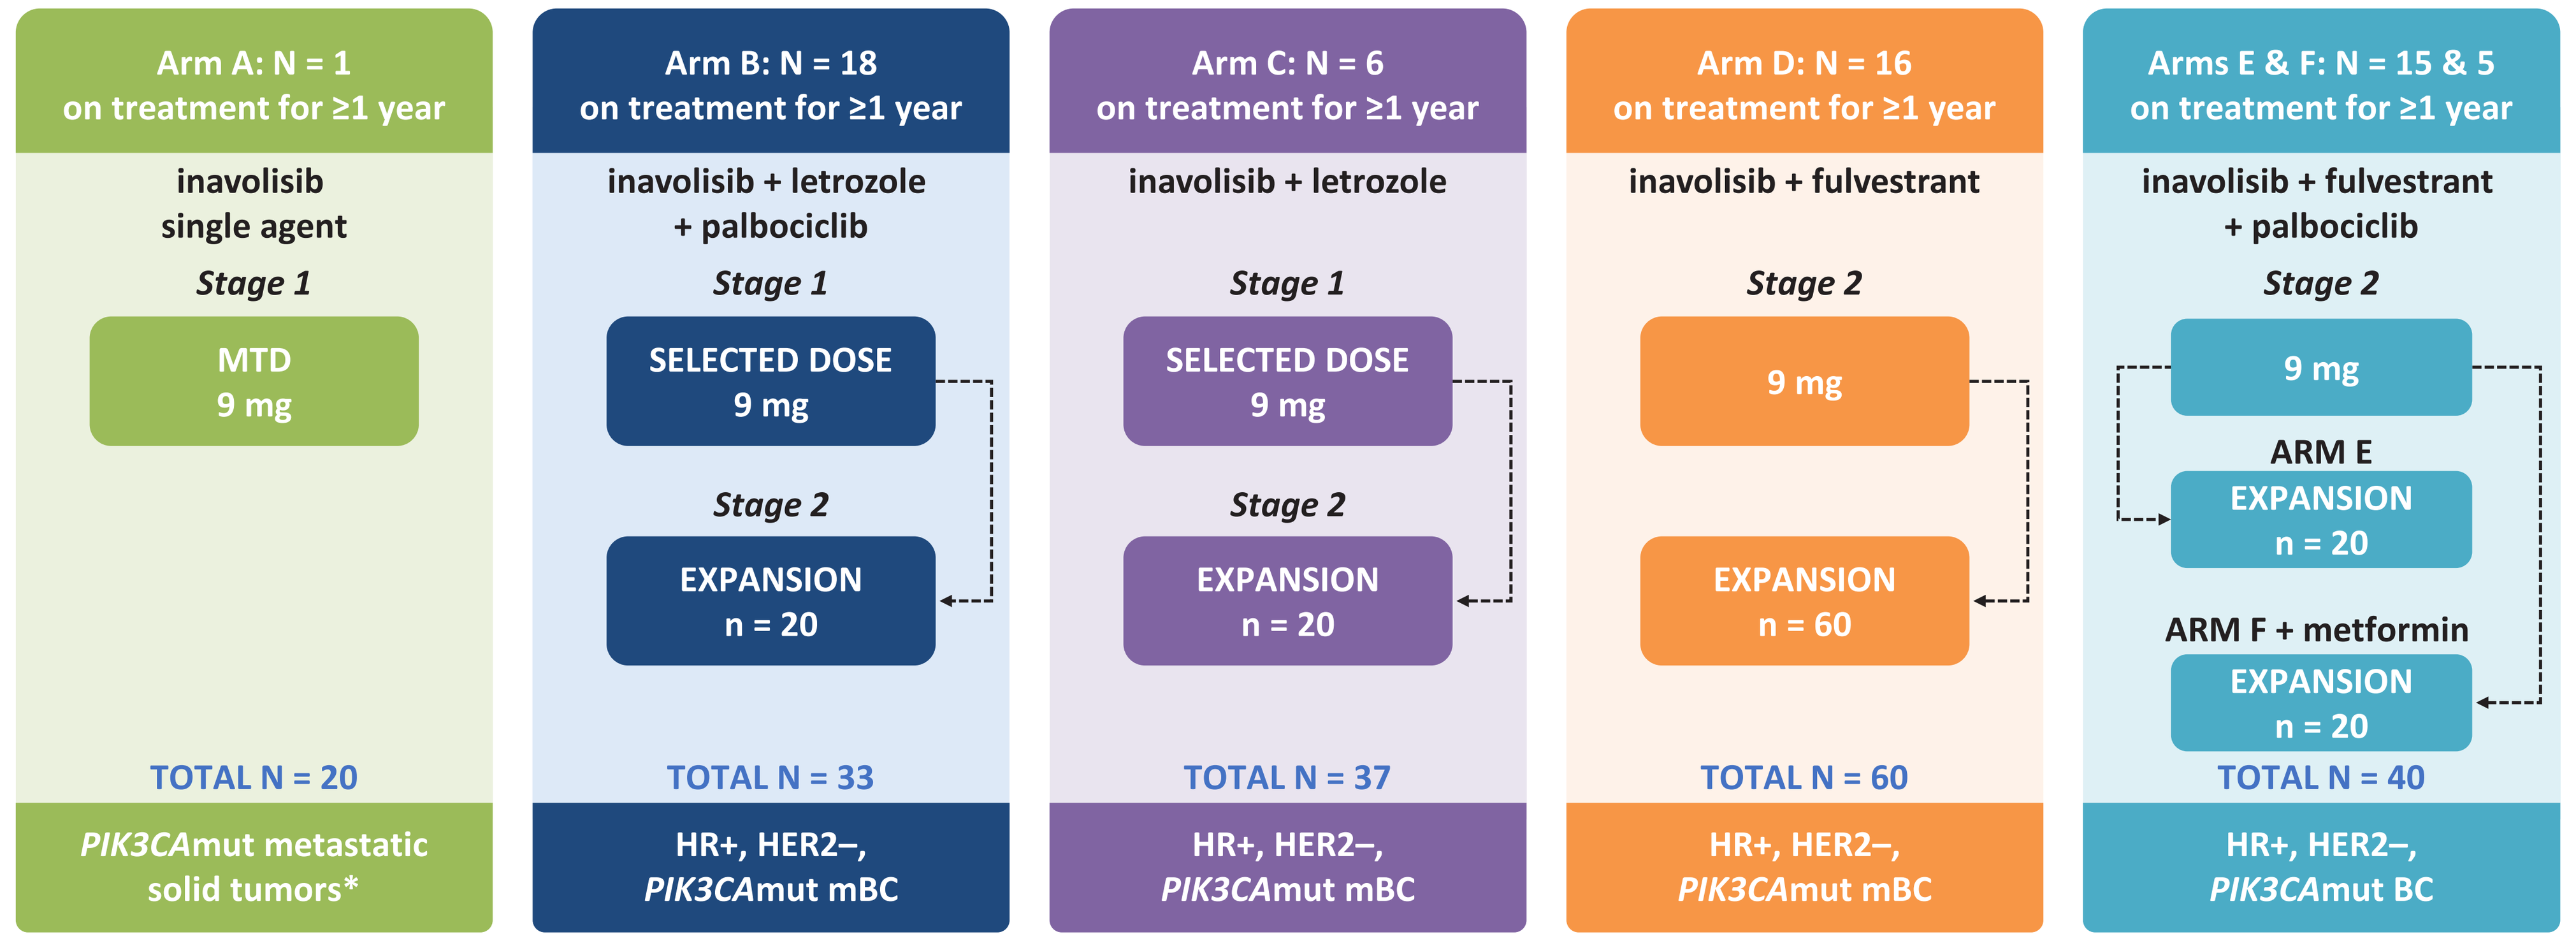
Arm A: N = 1
on treatment for ≥1 year
inavolisib
single agent
Stage 1
MTD
9 mg
TOTAL N = 20
PIK3CAmut metastatic
solid tumors*
Arm B: N = 18
on treatment for ≥1 year
inavolisib + letrozole
+ palbociclib
Stage 1
SELECTED DOSE
9 mg
Stage 2
EXPANSION
n = 20
TOTAL N = 33
HR+, HER2–,
PIK3CAmut mBC
Arm C: N = 6
on treatment for ≥1 year
inavolisib + letrozole
Stage 1
SELECTED DOSE
9 mg
Stage 2
EXPANSION
n = 20
TOTAL N = 37
HR+, HER2–,
PIK3CAmut mBC
Arm D: N = 16
on treatment for ≥1 year
inavolisib + fulvestrant
Stage 2
9 mg
EXPANSION
n = 60
TOTAL N = 60
HR+, HER2–,
PIK3CAmut mBC
Arms E & F: N = 15 & 5
on treatment for ≥1 year
inavolisib + fulvestrant
+ palbociclib
Stage 2
9 mg
ARM E
EXPANSION
n = 20
ARM F + metformin
EXPANSION
n = 20
TOTAL N = 40
HR+, HER2–,
PIK3CAmut BC

- Fasting glucose ≤140 mg/dL, HbA1c <7%; Arm F: BMI ≥30 kg/m^2 and/or HbA1c ≥5.7%.
- Inavolisib was administered PO QD at 3, 6, 9, or 12 mg (3+3 dose-escalation design; inavolisib dose expansion was 9 mg).
- Letrozole was administered at 2.5 mg PO QD.
- Palbociclib was administered at 125 mg PO QD for 21/28 days.
- Fulvestrant was administered at 500 mg intramuscularly every 4 weeks.
- Patients were administered study treatment in 28-day cycles until intolerable toxicity or disease progression.
- Safety was assessed by National Cancer Institute Common Terminology Criteria for Adverse Events v4.

Adapted from Kalinsky K, *et al.*[6]
Arm G (inavolisib + pertuzumab + trastuzumab on 21-day cycles) in HER2+ mBC is not depicted here, since no patients were part of the long-term safety population (three patients had been enrolled in the study overall at data cutoff date).
* All but one patient had breast cancer: one patient had colorectal cancer, but was not part of the long-term safety population.
BMI, body mass index; HbA1c, glycosylated hemoglobin; HR, hormone receptor; mBC, metastatic breast cancer; MTD, maximum tolerated dose; mut, mutated; PO, oral; QD, daily.

RESULTS

Patients

- The data cutoff date for this analysis was January 1, 2022; 193 patients were included in the overall safety population, and 61 were part of the long-term safety population (≥1 year to ~4 years). Baseline characteristics and prior cancer treatments are shown in Table 1.

Safety

- The safety profile of long-term treatment vs all enrolled patients was consistent, with no differences in the safety profiles and no new safety signals observed (Table 2).
- The most common treatment-related adverse events (AEs) were hyperglycemia, stomatitis, diarrhea, asthenia/fatigue, and nausea. The vast majority (>80%) of neutropenia events occurred in the palbociclib-containing arms (all Grade 3/4 neutropenia events occurred in the palbociclib-containing arms).
- Two patients experienced pneumonitis overall, both assessed as treatment-related and in palbociclib-containing arms; one Grade 1 in the long-term safety population.
- There were seven Grade 5 AEs reported overall (all unrelated to study treatment), with one in the long-term treatment group (pleural effusion; disease progression-related).
- One-hundred patients (52%) overall had ≥1 inavolisib-related AE resulting in study treatment modification (drug interruption/dose reduction/drug withdrawal); 34 (18%) had a dose reduction, 80 (41%) had an interruption, and four (2%) had treatment withdrawn due to an AE.
- Thirty-seven patients (61%) in the long-term treatment group had ≥1 inavolisib-related AE resulting in study treatment modification (drug interruption/dose reduction/drug withdrawal); nine (15%) had a dose reduction, 34 (56%) had an interruption, and one (2%) had treatment withdrawn due to an AE. Study drug modifications for targeted related AEs in this group are shown in Table 3.
- The most common medications for management of targeted AEs are shown in Table 4. Medications were not required for rash in the long-term patient population.
- Frequency of hyperglycemia, diarrhea, and stomatitis (grouped term including stomatitis, mucosal inflammation, mouth ulceration, glossodynia, lip ulceration, palatal ulcer, and tongue ulceration) decreased over time; rash (grouped term including rash, rash maculopapular, dermatitis acneiform, hand dermatitis, and rash pruritic) also decreased or had a stable frequency over time (Figure 2).
- Numerical comparisons between populations should be made with caution due to length of treatment and sample size differences.

Table 1: Patient characteristics and treatment exposure

	All patients N = 193	Patients on treatment ≥1 year N = 61
Median age, years (range)	59 (31–85)	58 (33–80)
BMI ≥30 kg/m^2, n/N (%)	50/191 (26%)	19/60 (32%)
HbA1c ≥5.7% to <7%, n/N (%)	62/192 (32%)	18/60 (30%)
ECOG PS 0, n (%)*	112 (58%)	36 (59%)
One prior chemotherapy for mBC, n/N (%)	24/87 (28%)	9/20 (15%)
≥2 prior lines of chemotherapy for mBC, n/N (%)	63/87 (72%)	11/20 (55%)
Median prior lines of therapy for mBC, n (range)	2 (1–10)	2 (1–7)
Prior fulvestrant, n (%)	96 (50%)	22 (36%)
Prior CDK4/6i, n (%)	125 (65%)	24 (39%)
Median inavolisib treatment duration, months (range)	6.4 (0.2–46.2)	20.6 (11.9–46.2)
Median cumulative inavolisib dose intensity, %	96%	95%

* Eligibility for enrollment: ECOG PS 0 or 1.
BMI, body mass index; CDK4/6i, cyclin-dependent kinase 4/6 inhibitor; ECOG PS, Eastern Cooperative Oncology Group performance status; HbA1c, glycosylated hemoglobin; mBC, metastatic breast cancer.

Table 2: Most common treatment-related* AEs (any grade in >10% of patients on treatment ≥1 year)

	All patients N = 193		Patients on treatment ≥1 year N = 61	
	Any grade	Grade ≥3	Any grade	Grade ≥3
Total number of patients with ≥1 AE	183 (95%)	104 (54%)	61 (100%)	44 (72%)
Hyperglycemia	125 (65%)	42 (22%)	43 (70%)	10 (16%)
Stomatitis (grouped term)†	84 (44%)	6 (3%)	40 (66%)	2 (3%)
Diarrhea	81 (42%)	1 (1%)	30 (49%)	1 (2%)
Nausea	67 (35%)	3 (2%)	23 (38%)	0
Neutropenia	58 (30%)	45 (23%)	33 (54%)	28 (46%)
Headache	14 (7%)	0	8 (13%)	0
Vomiting	31 (16%)	1 (1%)	10 (16%)	0
Anemia	34 (18%)	5 (3%)	19 (31%)	1 (2%)
Thrombocytopenia	33 (17%)	7 (4%)	21 (34%)	5 (8%)
Decreased appetite	46 (24%)	1 (1%)	15 (25%)	0
Abdominal pain	16 (8%)	0	8 (13%)	0
Muscle spasms	18 (9%)	0	9 (15%)	0
Dysgeusia	38 (20%)	0	16 (26%)	0
Fatigue	40 (21%)	6 (3%)	13 (21%)	1 (2%)
Asthenia	22 (11%)	1 (1%)	11 (18%)	0
Rash (grouped term)‡	27 (14%)	1 (1%)	11 (18%)	1 (2%)
Dry skin	13 (7%)	0	9 (15%)	0
Gastroesophageal reflux disease	14 (7%)	0	7 (11%)	0
Leukopenia	18 (9%)	8 (4%)	10 (16%)	5 (8%)
Lymphopenia	16 (8%)	5 (3%)	10 (16%)	3 (5%)
Dry mouth	21 (11%)	1 (1%)	7 (11%)	1 (2%)

Data are number of patients (%). * Does not include AEs related to metformin (patients in Arm F). † Stomatitis, mucosal inflammation, mouth ulceration, glossodynia, lip ulceration, palatal ulcer, tongue ulceration. ‡ Rash, rash maculopapular, dermatitis acneiform, hand dermatitis, rash pruritic.
AE, adverse event.

Table 3: Study drug modifications for targeted AEs related to inavolisib, in patients on treatment ≥1 year

	N = 61
Overall	37 (61%)
Dose reduced	9 (15%)
Drug interrupted	34 (56%)
Drug withdrawn	1 (2%)
Hyperglycemia	21 (34%)
Dose reduced	1 (2%)
Drug interrupted	20 (33%)
Drug withdrawn	0
Diarrhea	4 (7%)
Dose reduced	2 (3%)
Drug interrupted	3 (5%)
Drug withdrawn	1 (2%)
Rash (grouped term)*	2 (3%)
Dose reduced	0
Drug interrupted	2 (3%)
Drug withdrawn	0
Stomatitis (grouped term)†	6 (10%)
Dose reduced	3 (5%)
Drug interrupted	4 (7%)
Drug withdrawn	0

Table 4: Most common treatments (in ≥10% of patients) for targeted AEs in patients on treatment ≥1 year

	N = 61
Total number of patients with ≥1 treatment	53 (87%)
Hyperglycemia	**30 (49%)**
Metformin	27 (44%)
Sitagliptin	12 (20%)
Empagliflozin	11 (18%)
Diarrhea	**27 (44%)**
Loperamide	22 (36%)
Stomatitis (grouped term)†	**34 (56%)**
Dexamethasone mouthwash	27 (44%)
Lidocaine	8 (13%)

Data in Tables 3 and 4 are number of patients (%). * Rash, rash maculopapular, dermatitis acneiform, hand dermatitis, rash pruritic. † Stomatitis, mucosal inflammation, mouth ulceration, glossodynia, lip ulceration, palatal ulcer, tongue ulceration.
AE, adverse event.

Figure 2: Number of targeted AEs over time in patients on treatment ≥1 year

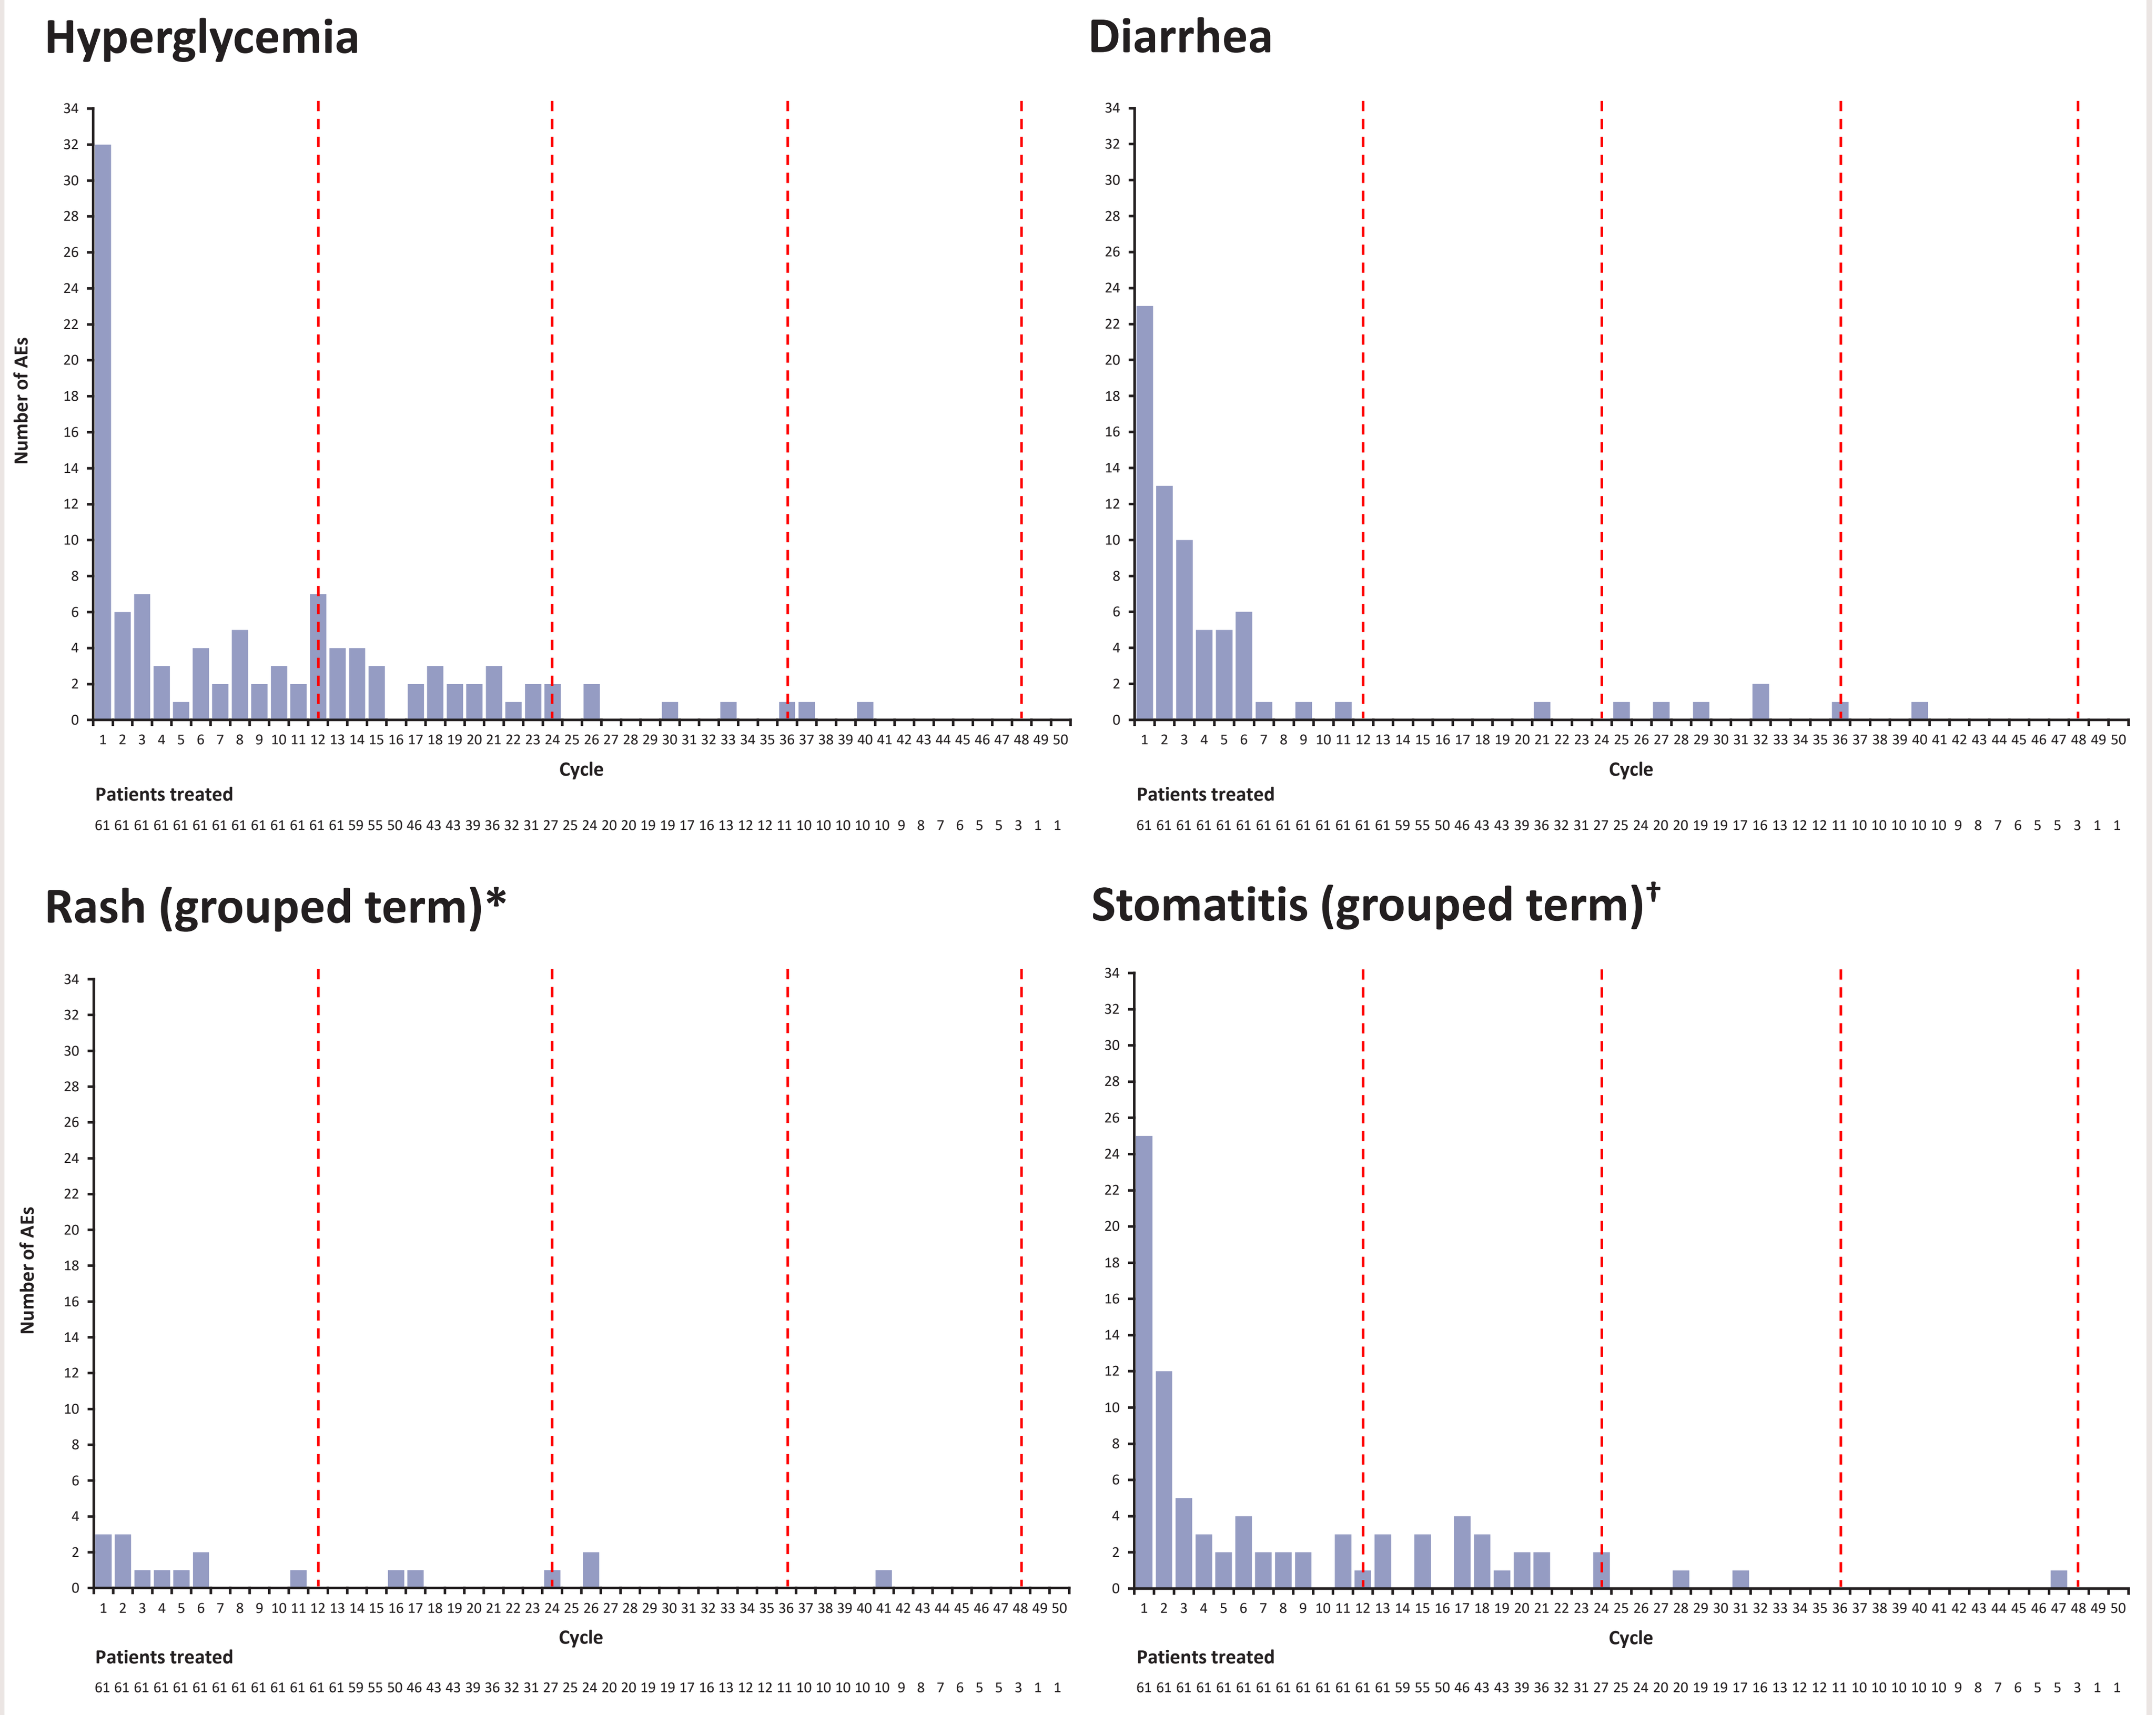
Hyperglycemia
Diarrhea
Rash (grouped term)*
Stomatitis (grouped term)†
Number of AEs
Cycle
Patients treated
61 61 61 61 61 61 61 61 61 61 61 61 59 55 50 46 43 43 39 36 32 31 27 25 24 20 20 19 19 17 16 13 12 12 11 10 10 10 10 10 9 8 7 6 5 5 3 1 1

Red vertical dashed lines represent years. * Rash, rash maculopapular, dermatitis acneiform, hand dermatitis, rash pruritic. † Stomatitis, mucosal inflammation, mouth ulceration, glossodynia, lip ulceration, palatal ulcer, tongue ulceration.
AE, adverse event.

CONCLUSIONS

- **These data indicate encouraging long-term safety and tolerability of inavolisib, and no new or unexpected safety signals were identified.**
- **The safety profile of patients on study treatment with inavolisib alone or in combination with endocrine-based anticancer therapies for ≥1 year was similar to that reported for the overall study population.**
- **The inavolisib development program is expanding, including a phase 3 study of inavolisib + palbociclib + fulvestrant that is currently enrolling (NCT04191499; INAVO120).[11]**

REFERENCES

1. Saal LH, *et al. Cancer Res* 2005; **65:**2554–2559; 2. Musgrove EA & Sutherland RL. *Nat Rev Cancer* 2009; **9:**631–643; 3. Stemke-Hale K, *et al. Cancer Res* 2008; **68:** 6084–6091; 4. Juric D, *et al.* SABCS 2019; OT1-08-04; 5. Jhaveri K, *et al.* SABCS 2019; P1-19-46; 6. Kalinsky K, *et al.* AACR 2020; 10349; 7. Bedard PL, *et al*. SABCS 2020; PD1-02; 8. Oliveria M, *et al*. SABCS 2020; PS11-11; 9. Jhaveri K, *et al*. SABCS 2020; PS5-12; 10. Juric D, *et al*. SABCS 2021; P5-17-05; 11. Turner N, *et al.* ESMO 2020; 355TiP.

ACKNOWLEDGMENTS

We thank the patients and their families who took part in the study, as well as the staff, research coordinators, and investigators at each participating institution. Pfizer, Inc. provided palbociclib for this study. Support for third-party writing assistance for this poster, furnished by Daniel Clyde, PhD, of Health Interactions, was provided by F. Hoffmann-La Roche Ltd.

CONFLICTS OF INTEREST

PB reports consulting or advisory roles for Bristol Myers Squibb and Pfizer (paid to his institution) and for Seattle Genetics, Lilly, Amgen, Merck, and Gilead Sciences (personal); and research funding from Bristol-Myers-Squibb, Sanofi, AstraZeneca, Genentech/Roche, SERVIER, GlaxoSmithKline, Novartis, PTC Therapeutics, Nektar, Merck, Seattle Genetics, Mersana, Immunomedics, Lilly, Amgen, and Bicara Therapeutics (paid to his institution). Please refer to the abstract for all author conflicts of interest. All authors have received research support in the form of third-party writing assistance for this poster from F. Hoffmann-La Roche Ltd. This study is sponsored by Genentech, Inc.

This presentation is the intellectual property of the authors. Contact philippe.bedard@uhn.ca for permission to reprint and/or distribute.