San Antonio Breast Cancer Symposium® – December 7–10, 2021

A phase I/Ib study of inavolisib (GDC-0077) in combination with fulvestrant in patients (pts) with *PIK3CA*-mutated hormone receptor-positive/HER2-negative (HR+/HER2–) metastatic breast cancer

P5-17-05

Dejan Juric[1], Philippe L Bedard[2], Andrés Cervantes[3], Valentina Gambardella[3], Mafalda Oliveira[4], Cristina Saura[4], Kevin M Kalinsky[5], Erika Hamilton[6], Antoine Italiano[7], Ian E Krop[8], Peter Schmid[9], Nicholas C Turner[10], Andrea Varga[11], Stephanie Royer-Joo[12], Katherine E Hutchinson[12], Jennifer L Schutzman[12], Komal L Jhaveri[13]

[1]Massachusetts General Hospital, Boston, MA; [2]University of Toronto, Toronto, ON, Canada; [3]Biomedical Research Institute INCLIVA, University of Valencia, Valencia, Spain; [4]Vall d'Hebron University Hospital and Vall d'Hebron Institute of Oncology (VHIO), Barcelona, Spain; [5]Columbia University Irving Medical Center, New York, NY; [6]Sarah Cannon Research Institute/Tennessee Oncology, Nashville, TN; [7]Institut Bergonié, Bordeaux, France; [8]Dana-Farber Cancer Institute, Boston, MA; [9]Centre for Experimental Cancer Medicine, Cancer Research UK Barts Centre, London, UK; [10]Royal Marsden Hospital and Institute of Cancer Research, London, UK; [11]Gustave Roussy Cancer Campus, Villejuif, France; [12]Genentech, Inc., South San Francisco, CA; [13]Memorial Sloan Kettering Cancer Center, New York, NY

BACKGROUND

- Dysregulating mutations in *PIK3CA*, which encodes the catalytic PI3K p110a subunit, are common in breast cancer and other solid tumors;[1–3] however, there are only limited data available on the role of PI3Ka inhibition in the post-cyclin-dependent kinase 4/6 inhibitor (CDK4/6i) setting.
- Inavolisib is a PI3Ka-selective inhibitor and degrader of mutated PI3Ka that has demonstrated encouraging preliminary antitumor activity in patients with *PIK3CA*-mutated HR+ breast cancer as a single agent, and in combination with antiestrogen therapy.[4,5]
- An open-label, phase I/Ib dose-escalation study of inavolisib alone and in combination with endocrine and targeted therapies is ongoing (NCT03006172; GO39374); data for inavolisib in combination with fulvestrant in female pts with *PIK3CA*-mutated, HR+/HER2– metastatic breast cancer (Arm D) are presented.

METHODS

- The study design for Arm D of GO39374, including key eligibility criteria and endpoints, is shown in Figure 1.

Figure 1: Study design (Arm D), key eligibility criteria, and key endpoints

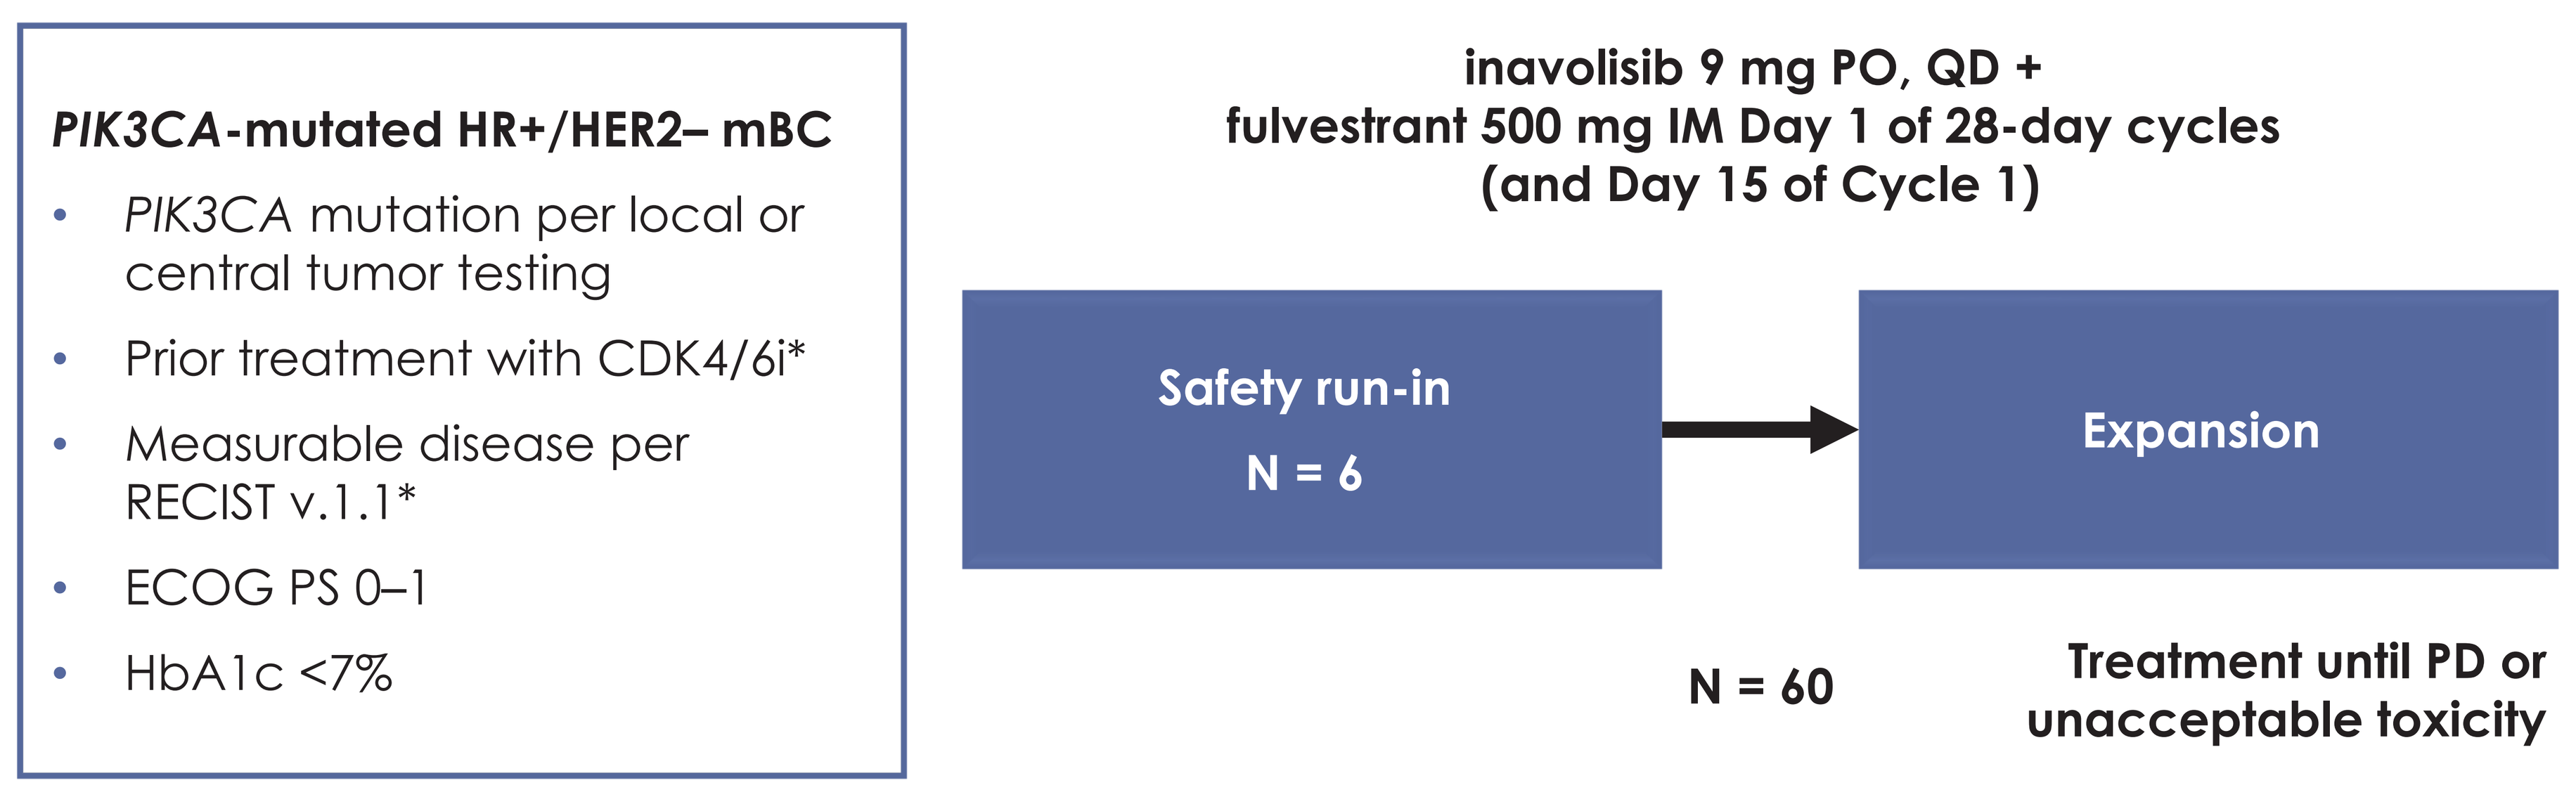
PIK3CA-mutated HR+/HER2– mBC
PIK3CA mutation per local or central tumor testing
Prior treatment with CDK4/6i*
Measurable disease per RECIST v.1.1*
ECOG PS 0–1
HbA1c <7%
inavolisib 9 mg PO, QD + fulvestrant 500 mg IM Day 1 of 28-day cycles (and Day 15 of Cycle 1)
Safety run-in
N = 6
Expansion
N = 60
Treatment until PD or unacceptable toxicity

- Pts who were pre-/perimenopausal were treated with gonadotropin-releasing hormone or luteinizing hormone-releasing hormone agonist therapy.
- *PIK3CA* mutant allele frequency was assessed in ctDNA from serial plasma collections using FoundationACT™.
- Endpoints include safety (NCI-CTCAE v4); PK; preliminary antitumor activity (assessed every 8 weeks via RECIST v1.1; CBR: stable disease for ≥24 weeks, PR, or CR; PFS); and signaling and pharmacodynamic biomarkers using ctDNA.

* For the initial study protocol (first 20 pts), prior treatment with a CDK4/6i and measurable disease per RECIST v.1.1 were not required (disease could be evaluable per RECIST v.1.1). CBR, clinical benefit rate; CDK4/6i, cyclin-dependent kinase 4/6 inhibitor; CR, complete response; ctDNA, circulating tumor DNA; ECOG PS, Eastern Cooperative Oncology Group performance status; HbA1c, glycated hemoglobin; HR, hormone receptor; IM, intramuscular; mBC, metastatic breast cancer; NCI-CTCAE, National Cancer Institute Common Terminology Criteria for Adverse Events; PD, disease progression; PFS, progression-free survival; PK, pharmacokinetics; PO, oral; PR, partial response; pt, patient; QD, daily; RECIST, Response Evaluation Criteria in Solid Tumors.

RESULTS

Patients

- The data cut-off date for this analysis was July 26, 2021; 60 pts were enrolled; 19 were still on study treatment at the time of data cut-off.
- Baseline characteristics are shown in Table 1.
- All but two pts had received a prior CDK4/6i before study treatment.
- Forty-one pts (68%) discontinued inavolisib; 36 due to radiographic progression, three due to symptomatic deterioration, one due to physician's decision, and one due to death (unrelated serious adverse event of Grade 5 hypertrophic cardiomyopathy on Study Day 8).

Safety

- Common adverse events are shown in Table 2.
- Thirty-five pts (58%) had an adverse event leading to dose modification. Six pts (10%) required dose reductions. No treatment-related adverse event resulted in treatment withdrawal.
 - Dose modifications included withdrawals, reductions, and interruptions. Reductions may have resulted in interruptions prior to reduction.
- Hyperglycemia was managed with antihyperglycemic agents in 23 pts (38%), and with inavolisib dose modifications in 21 pts (35%).
- Stomatitis (grouped term; defined as stomatitis, mucosal inflammation, and mouth ulceration) was reported in 15 pts (25%) and managed in the vast majority of cases with dexamethasone mouthwash.
- Rash (grouped term; defined as rash and rash maculo-papular) was reported in seven pts (12%). All rash events were Grade 1.

Table 1: Pt characteristics and treatment exposure

	N = 60
Median age, years (range)	59.5 (31–85)
BMI ≥30 kg/m², n (%)	9 (15%)
HbA1c ≥5.7%, n (%)	19 (32%)
ECOG PS 0, n (%)	34 (57%)
One prior chemotherapy for mBC, n (%)	25 (42%)
≥2 prior lines of therapy for mBC, n (%)	34 (57%)
Median prior lines of therapy for mBC, n (range)	2 (1–7)
Prior fulvestrant, n (%)	28 (47%)
Prior CDK4/6i, n (%)*	58 (97%)
Median inavolisib treatment duration, months (range)	5.5 (0.2–32.6)
Median cumulative inavolisib dose intensity, %	98%
Median fulvestrant treatment duration, months (range)	4.7 (0–31.8)
Median cumulative fulvestrant dose intensity, %	100%

* Two pts were enrolled without prior CDK4/6i when not required in the initial protocol design (one pt treated with letrozole alone in the metastatic setting and one pt with heterogeneous cancer prior to study entry who received prior treatment with trastuzumab).
BMI, body mass index; CDK4/6i, cyclin-dependent kinase 4/6 inhibitor; ECOG PS, Eastern Cooperative Oncology Group performance status; HbA1c, glycated hemoglobin; mBC, metastatic breast cancer; pt, patient.

Table 2: Common treatment-related AEs (≥10% of pts at any grade)

	N = 60	
	Any grade	**Grade ≥ 3**
Pts with ≥1 AE	56 (93%)	20 (33%)
Hyperglycemia	37 (62%)	13 (22%)
Diarrhea	25 (42%)	0
Nausea	19 (32%)	2 (3%)
Stomatitis*	15 (25%)	0
Decreased appetite	10 (17%)	0
Dysgeusia	10 (17%)	0
Fatigue	8 (13%)	0
Asthenia	8 (13%)	0
Muscle spasms	7 (12%)	0
Rash†	7 (12%)	0
Alopecia	6 (10%)	0
Mucosal inflammation	6 (10%)	0

Data are number of pts (%).
* Grouped terms: stomatitis, mucosal inflammation, and mouth ulceration.
† Grouped terms: rash and rash maculo-papular.
AE, adverse event; pt, patient.

Pharmacodynamics

- At clinical cut-off, circulating tumor (ct)DNA-derived *PIK3CA* mutation data were available for 30/60 enrolled pts.
- Lack of data is due either to pt samples not being sequenced by the time of clinical cut-off, or to sequencing failure.
- In the majority of pts with paired *PIK3CA* mutation results, allele frequencies decreased between the Cycle 1, Day 1 and Cycle 1, Day 15 timepoints (Figure 2).

Figure 2: ctDNA *PIK3CA* MAF between Cycle 1, Day 1 and Cycle 1, Day 15

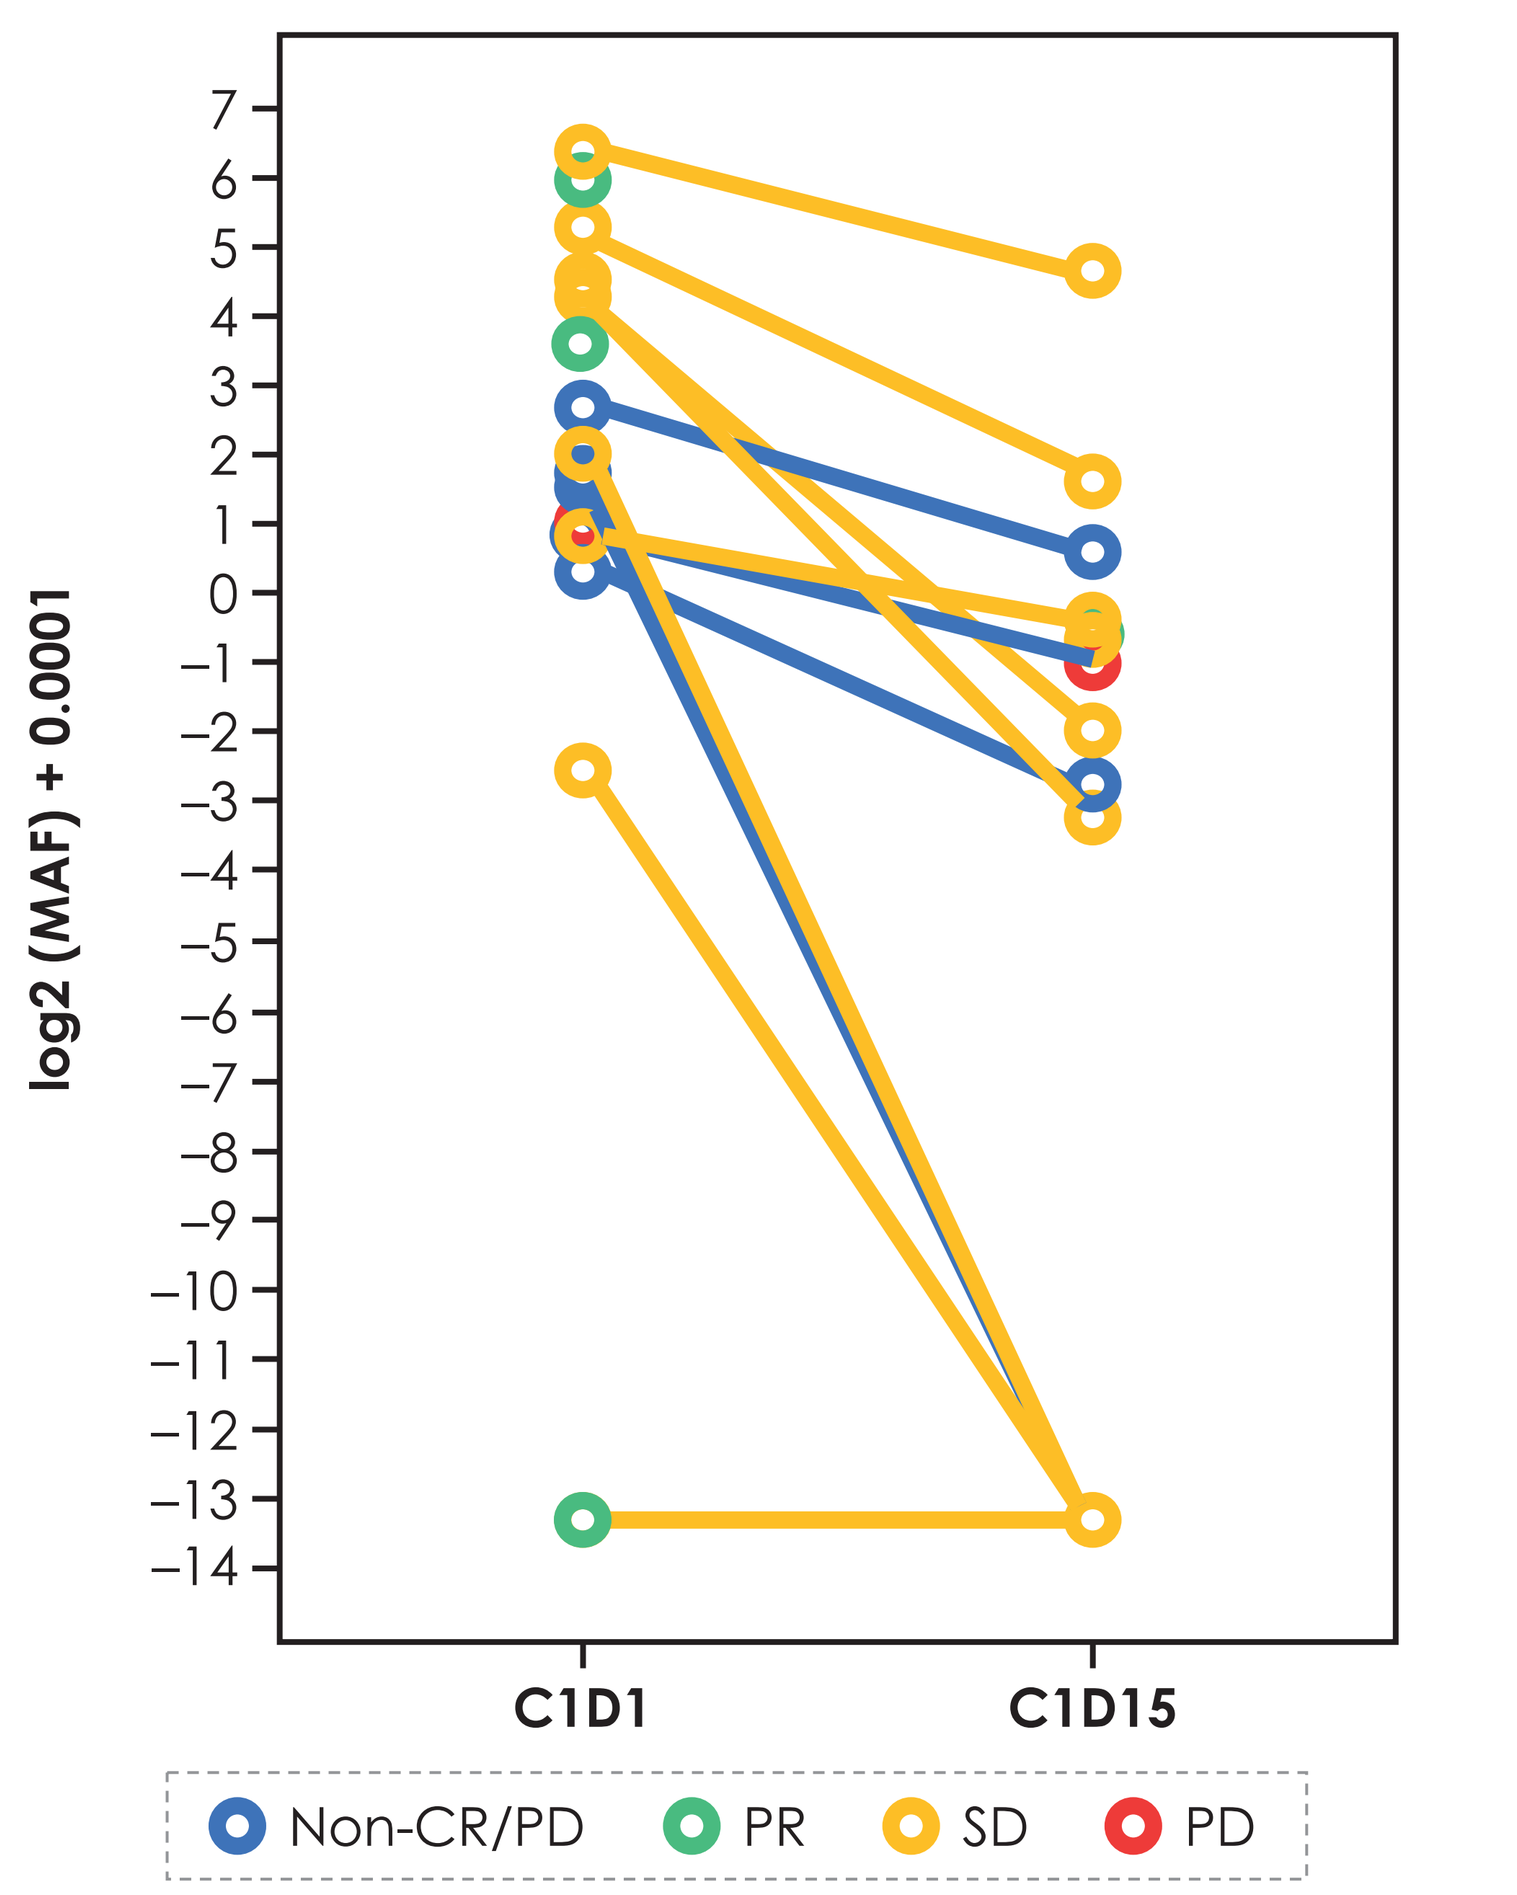
log2 (MAF) + 0.0001
7
6
5
4
3
2
1
0
−1
−2
−3
−4
−5
−6
−7
−8
−9
−10
−11
−12
−13
−14
C1D1
C1D15
Non-CR/PD
PR
SD
PD

C, cycle; CR, complete response; ctDNA, circulating tumor DNA; D, day; MAF, mutant allele frequency; PD, disease progression; SD, stable disease.

Pharmacokinetics

- The pharmacokinetics (PK) of inavolisib in combination with fulvestrant was similar to single-agent inavolisib PK.
- Similarly, the PK of fulvestrant in combination with inavolisib in this study was comparable to historical fulvestrant PK.
- Taken together, the results suggest no drug–drug interactions between inavolisib and fulvestrant.

Clinical activity (Figure 3)

- Overall, 14/54 pts with measurable disease achieved a partial response (PR) (26%; four of the responding pts had received prior fulvestrant; 13, prior CDK4/6i). Ten pts (19%) had a confirmed PR.
- The clinical benefit rate was 48% (29/60 pts).
- Median progression-free survival was 7.1 months (0 [censored]–32).

Figure 3: Clinical activity

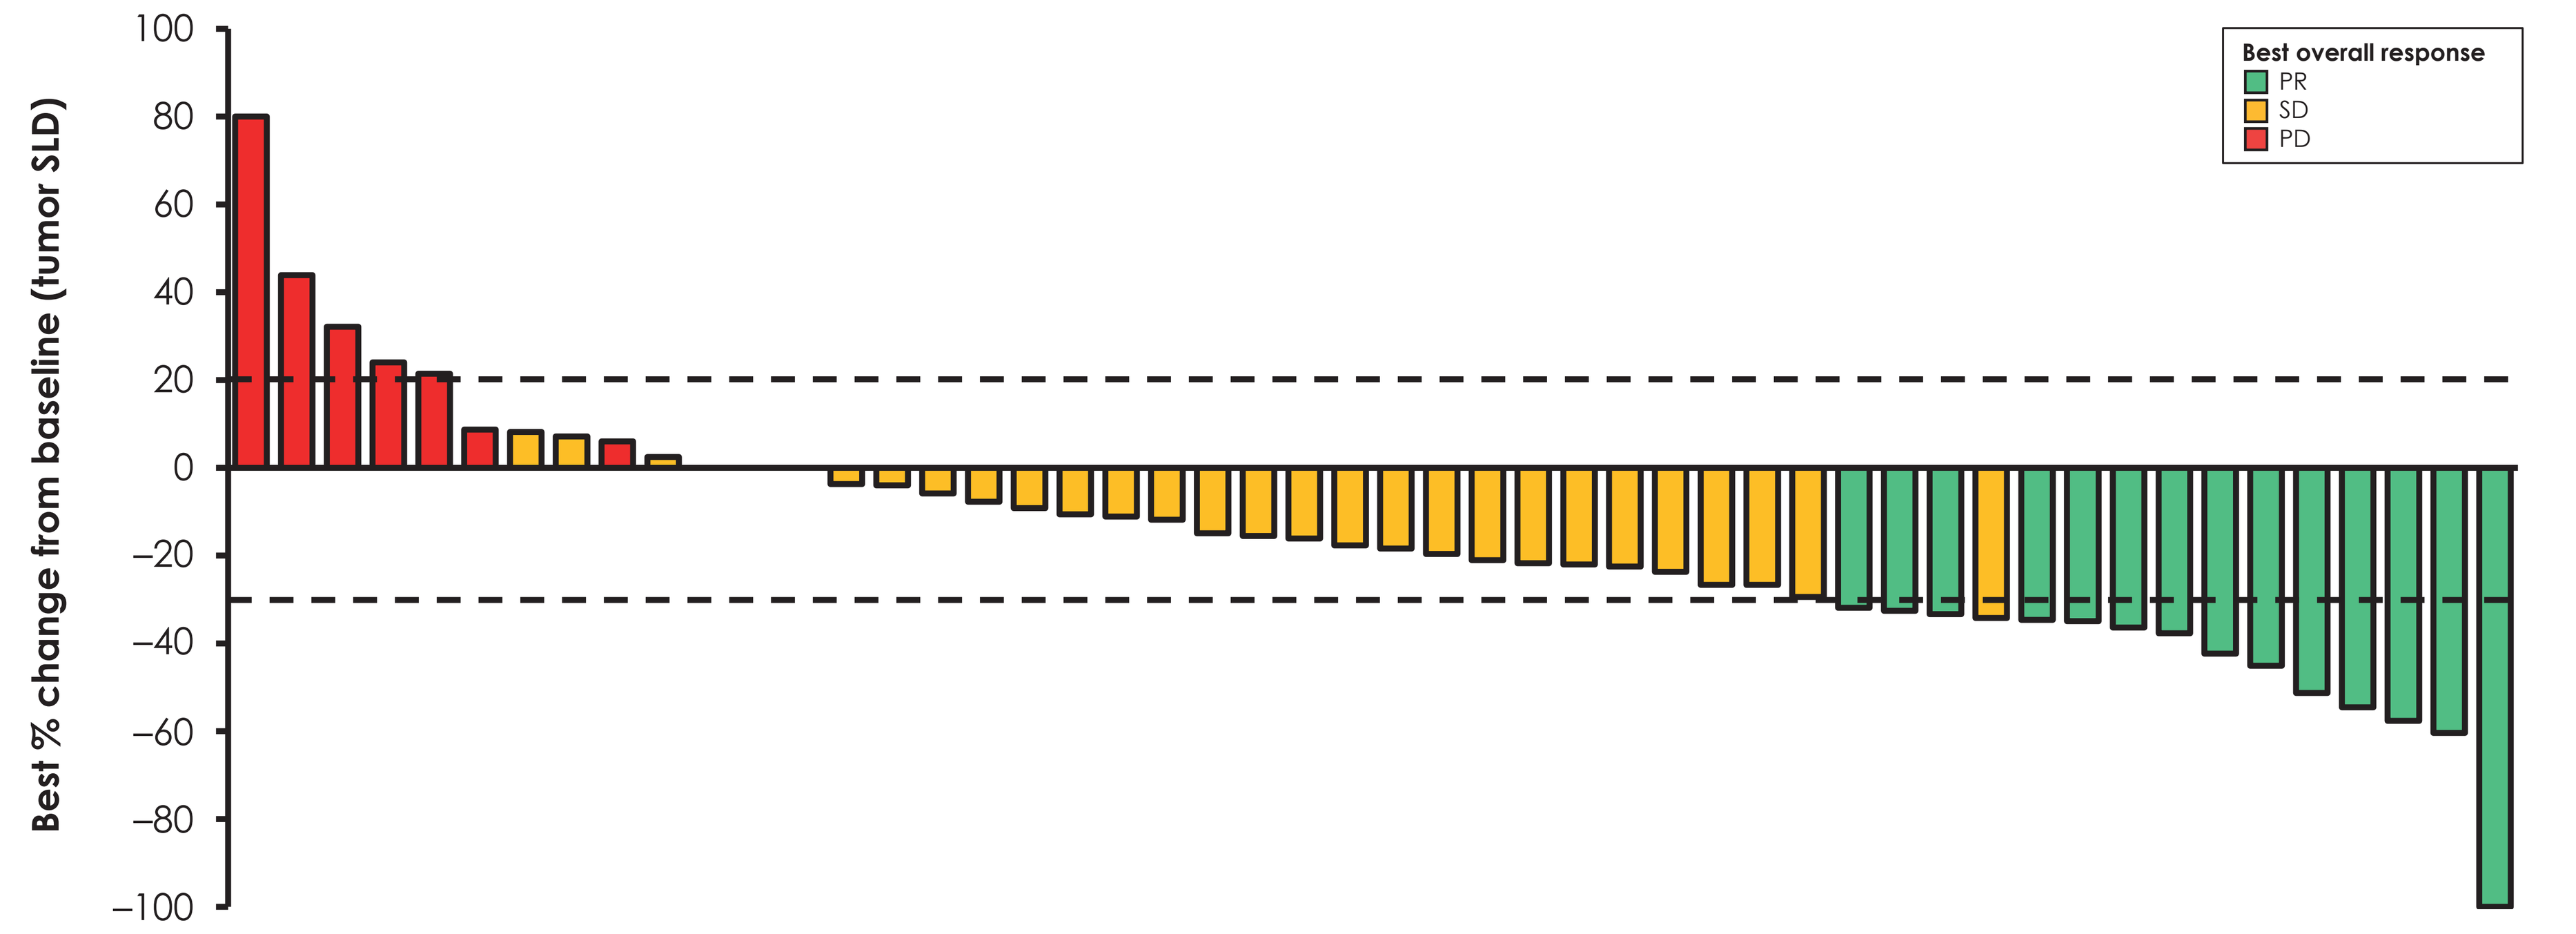
Best % change from baseline (tumor SLD)
100
80
60
40
20
0
−20
−40
−60
−80
−100
Best overall response
PR
SD
PD

	1	2	3	4	5	6	7	8	9	10	11	12	13	14	15	16	17	18	19	20	21	22	23	24	25
p110a mutation	KIN	HEL	HEL	KIN	HEL	KIN	HEL	HEL	HEL	KIN	HEL	HEL	KIN	HEL	O	HEL	HEL	KIN	KIN	HEL	HEL	HEL	KIN	KIN	KIN
Time on treatment (day)*	55	67	49	56	52	57	98	103	42	113	389	83	357	154	140	253	183	218	67	95	121	168	160	164	393
Active	N	N	N	N	N	N	N	Y	N	N	N	N	Y	Y	Y	N	Y	N	Y	N	N	N	N	N	Y
Best response	PD	PD	PD	PD	PD	PD	SD	SD	PD	SD	SD	SD	SD	SD	SD	SD	SD	SD	SD	SD	SD	SD	SD	SD	SD
Liver metastases	N	Y	Y	Y	Y	Y	Y	Y	Y	Y	N	N	Y	Y	Y	Y	Y	Y	Y	Y	N	Y	Y	N	N
Line of therapy	2	4	2	2	4	2	3	2	6	5	2	4	3	4	2	2	3	5	4	3		8	3	4	5
Prior CDK4/6i	N	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y
Prior AI	M	M	N	A	M	B	A	A	B	M	M	M	B	A	M	M	A	M	A	M	N	M	M	B	M
Prior fulvestrant	N	Y	Y	Y	N	N	Y	Y	Y	Y	N	Y	Y	Y	N	N	Y	Y	Y	N	N	Y	N	Y	Y

	26	27	28	29	30	31	32	33	34	35	36	37	38	39	40	41	42	43	44	45	46	47	48	49	50
p110a mutation	HEL	HEL	KIN	HEL	HEL	HEL	HEL	KIN	KIN	O	KIN	HEL	HEL	KIN	Mut	HEL	HEL	KIN	HEL	HEL	HEL	HEL	KIN	HEL	O
Time on treatment (day)*	59	279	193	337	59	147	168	727	257	418	227	586	371	116	65	175	251	482	169	168	491	224	546	280	222
Active	N	N	Y	N	N	Y	N	Y	Y	N	N	N	Y	N	N	N	Y	Y	Y	N	Y	Y	Y	N	N
Best response	SD	SD	SD	SD	SD	SD	SD	SD	SD	SD	PR†	PR	PR†	SD	PR†	PR†	PR	PR	PR	PR	PR	PR	PR	PR	PR
Liver metastases	Y	Y	N	Y	Y	Y	Y	N	N	Y	N	N	N	Y	Y	Y	N	N	N	Y	N	N	Y	N	N
Line of therapy	2	2	2	2	2	4	3	2	3	2	4	8	3	2	3	3	2	2	2	3	2	2	3	3	2
Prior CDK4/6i	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	Y	N	Y
Prior AI	B	A	B	M	A	N	M	M	M	A	M	M	M	M	B	M	M	M	M	M	B	M	A	M	M
Prior fulvestrant	N	Y	N	N	Y	Y	N	N	N	Y	Y	Y	N	N	N	N	N	N	Y	N	N	N	Y	N	N

* Colored boxes indicate treatment times of ≥6 months.
† Unconfirmed PR.
A, adjuvant setting; AI, aromatase inhibitor; B, both adjuvant and metastatic settings; HEL, helical domain (E545, E542, Q546); KIN, kinase domain (H1047, M1043); M, metastatic setting; Mul, multiple mutations; N, no; O, other (N345K, C420R); PD, disease progression; PR, partial response; SD, stable disease; SLD, sum of longest diameters; U, unknown; Y, yes.

CONCLUSIONS

- **Inavolisib in combination with fulvestrant demonstrated a manageable safety profile and encouraging preliminary antitumor activity, including in pts who previously progressed on a CDK4/6i.**
- **A PK profile similar to that of inavolisib alone, lack of drug–drug interactions, and pharmacodynamic modulation (i.e., decreased *PIK3CA* mutant allele frequency in ctDNA) were also observed.**
- **Inavolisib continues to be developed in breast cancer and other solid tumors.**
- **A phase III, randomized, double-blind, placebo-controlled study assessing the efficacy, safety, and PK of inavolisib or placebo with palbociclib + fulvestrant in pts with *PIK3CA*-mutated, HR+/HER2– locally advanced/mBC is ongoing (NCT04191499).[6]**

REFERENCES

1. Saal LH, *et al. Cancer Res* 2005; **65:**2554–2559; 2. Musgrove EA & Sutherland RL. *Nat Rev Cancer* 2009; **9:**631–643; 3. Stemke-Hale K, *et al. Cancer Res* 2008; **68:**6084–6091; 4. Juric D, *et al.* SABCS 2019; OT1-08-04; 5. Jhaveri K, *et al.* SABCS 2019; P1-19-46. 6. Turner N, *et al.* ESMO 2020; 355TiP.

ACKNOWLEDGMENTS

We thank the patients and their families who took part in the study, as well as the staff, research coordinators, and investigators at each participating institution. Support for third-party writing assistance for this poster, furnished by Daniel Clyde, PhD, of Health Interactions, was provided by F. Hoffmann-La Roche Ltd.

CONFLICTS OF INTEREST

DJ reports consulting fees (e.g., advisory boards) from Novartis, Genentech, Inc., EMD Serono, Eisai, Ipsen, Syros, Vibliome Therapeutics, Relay Therapeutics, MapKure, Petra Pharma, Silverback Therapeutics, and PIC Therapeutics; and contracted research from Novartis, Genentech, Inc., Eisai, EMD Serono, Pfizer, Syros, Takeda, Amgen, InventisBio, Dizal Pharma, Celgene, and Infinity Pharmaceuticals. Please refer to the abstract for all author conflicts of interest. All authors have received research support in the form of third-party writing assistance for this poster from F. Hoffmann-La Roche Ltd. This study is sponsored by Genentech, Inc.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from SABCS® and the author of this poster.

https://bit.ly/3bt3Nkr

This presentation is the intellectual property of the authors. Contact Juric.Dejan@mgh.harvard.edu for permission to reprint and/or distribute.